Elacestrant combinations with ribociclib and everolimus in patients with ER+/HER2- locally advanced or metastatic breast cancer (mBC): Update from ELEVATE, a phase (Ph) 1b/2, open-label, umbrella study

Presented at ASCO® 2025
May 30–June 3, 2025
Abstract # 1070

Hope S. Rugo,[1] Sara M. Tolaney,[2] Nancy Chan,[3] Erika Hamilton,[4] Marina Sharifi,[5] Kristine J. Rinn,[6] Wassim McHayleh,[7] Rinat Yerushalmi,[8] Neelima Vidula,[9] Joyce O'Shaughnessy,[10] Giuseppe Curigliano,[11] Javier Cortés,[12] Paula Muñoz Romero,[13] Denise Lepley,[14] Jennifer A. Crozier,[14] Tomer Wasserman,[14] Virginia Kaklamani[15]

[1]City of Hope Duarte Cancer Center, Duarte, CA, USA; [2]Dana-Farber Cancer Institute, Boston, MA, USA; [3]NYU Langone Health, New York, NY, USA; [4]Sarah Cannon Research Institute, Nashville, TN, USA; [5]University of Wisconsin, Madison, WI, USA; [6]Cancer Care Northwest, Spokane, WA, USA; [7]Adventhealth Cancer Institute, Orlando, FL, USA; [8]Rabin Medical Center, Petah Tikva, Israel; [9]Massachusetts General Hospital, Harvard Medical School, Boston, MA, USA; [10]Baylor University Medical Center, Texas Oncology, US Oncology, Dallas, TX, USA; [11]Istituto Europeo di Oncologia, IRCCS, and University of Milano, Milano, Italy; [12]International Breast Cancer Center (IBCC), Quironsalud Group, Barcelona, Spain; [13]Menarini Group, Florence, Italy; [14]Menarini Group, New York, NY, USA; [15]University of Texas Health Sciences Center San Antonio, San Antonio, TX, USA

BACKGROUND

- After first-line endocrine therapy (ET) plus a CDK4/6 inhibitor (CDK4/6i) in patients with ER+/HER2-, advanced or metastatic breast cancer, tumors develop resistance to endocrine therapies, leading to disease progression.[1] Resistance mechanisms include intrinsic alterations (in the PI3K/AKT/mTOR or cell cycle pathways)[2-8] or *ESR1* mutations that represent a type of acquired resistance that emerges in up to 50% of patients after initial ET in the metastatic setting.[2,9-15]
- Several trials have shown improved median PFS (mPFS) with the addition of second-line everolimus (mPFS of 3.6-6.8 months)[16,17] or a switch in CDK4/6i (mPFS of 5.3 mo).[18] Continuing SOC ET is limited by resistance to ET due to *ESR1* mutations.[2,9-11]
- In the phase 3 EMERALD trial, single-agent elacestrant significantly improved PFS versus SOC ET in patients with *ESR1*-mut tumors (HR=0.55; 95% CI: 0.39-0.77; p=0.0005) and in all patients (HR=0.70; 95% CI: 0.55-0.88; p=0.0018).[19]
 - In patients with ≥12 months of prior ET + CDK4/6i and *ESR1*-mutated tumors, mPFS with elacestrant was 8.6 months vs 1.9 months with SOC (HR=0.41; 95% CI: 0.26-0.63).[20]
- The rationale for combining elacestrant with PI3K/AKT/mTOR or CDK4/6 inhibitors is to overcome different resistance mechanisms.[21,22]
- Elacestrant in combination with PI3K/AKT/mTOR or CDK4/6 inhibitors have shown favorable safety and encouraging anti-tumor activity.
- This analysis reports updated phase 1b safety and preliminary efficacy for elacestrant in combination with everolimus or ribociclib.

METHODS

ELEVATE Study Design

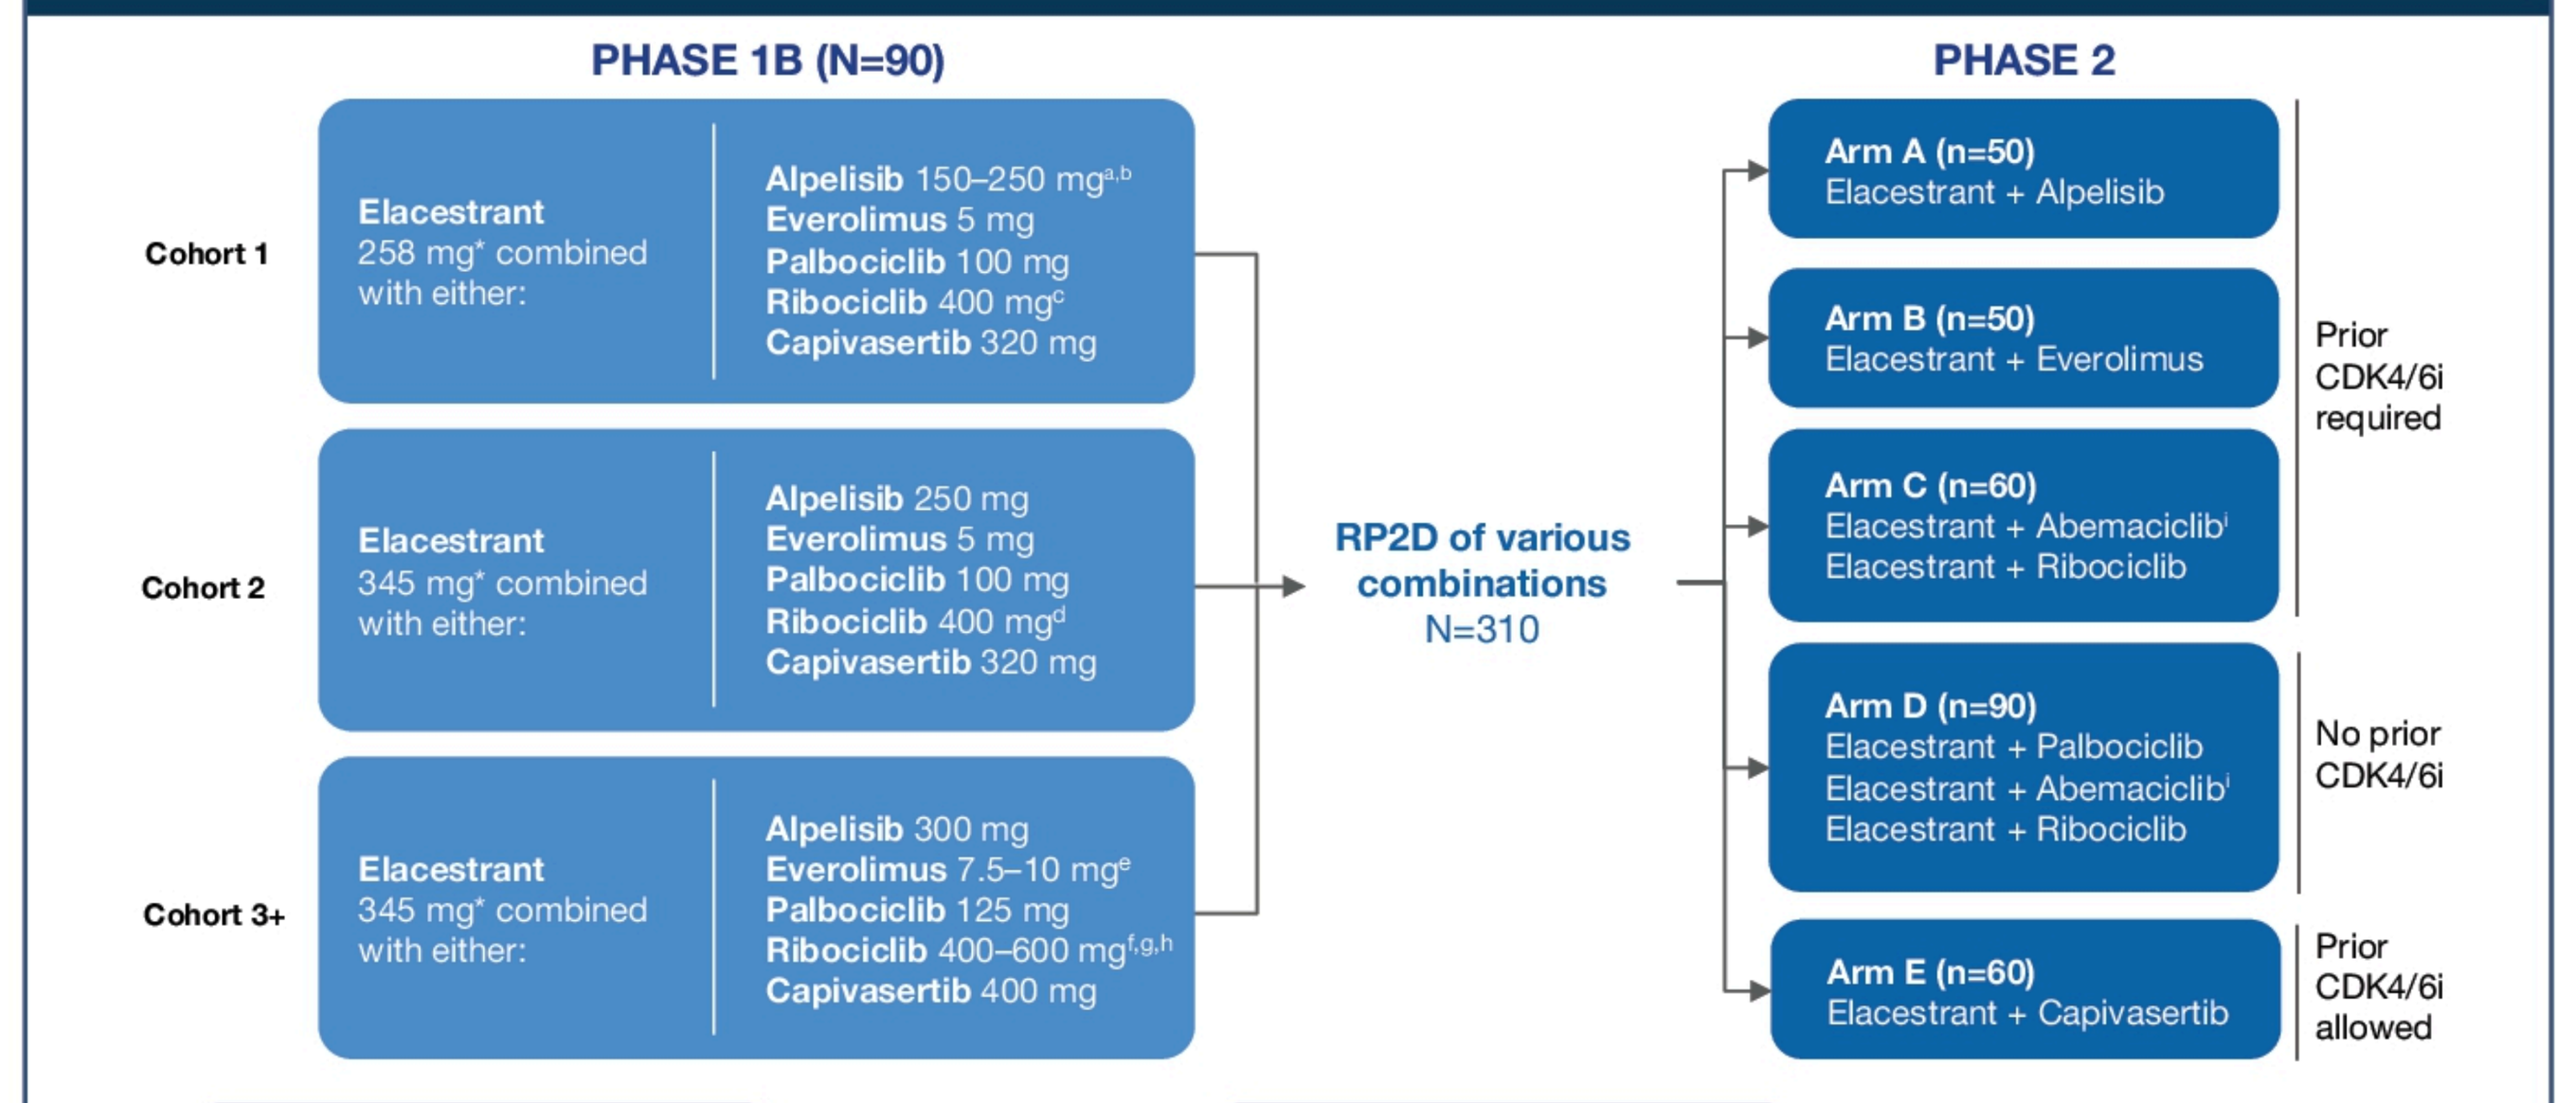
PHASE 1B (N=90)
PHASE 2
Cohort 1
Elacestrant 258 mg* combined with either:
Alpelisib 150–250 mg[a,b]
Everolimus 5 mg
Palbociclib 100 mg
Ribociclib 400 mg[c]
Capivasertib 320 mg
Cohort 2
Elacestrant 345 mg* combined with either:
Alpelisib 250 mg
Everolimus 5 mg
Palbociclib 100 mg
Ribociclib 400 mg[d]
Capivasertib 320 mg
Cohort 3+
Elacestrant 345 mg* combined with either:
Alpelisib 300 mg
Everolimus 7.5–10 mg[e]
Palbociclib 125 mg
Ribociclib 400–600 mg[f,g,h]
Capivasertib 400 mg
RP2D of various combinations
N=310
Arm A (n=50) Elacestrant + Alpelisib
Arm B (n=50) Elacestrant + Everolimus
Arm C (n=60) Elacestrant + Abemaciclib[i] Elacestrant + Ribociclib
Prior CDK4/6i required
Arm D (n=90) Elacestrant + Palbociclib Elacestrant + Abemaciclib[i] Elacestrant + Ribociclib
No prior CDK4/6i
Arm E (n=60) Elacestrant + Capivasertib
Prior CDK4/6i allowed

PHASE 1B OBJECTIVE

Primary: Determine RP2D of elacestrant in combination with each of the other study drugs

PHASE 2 OBJECTIVES

Primary: Evaluate the PFS of elacestrant in combination with each of the other study drugs

Secondary: Evaluate efficacy and safety of elacestrant in combination with each of the other study drugs

NCT05563220

Elacestrant 86 mg is equivalent to 100 mg elacestrant hydrochloride; elacestrant 172 mg is equivalent to 200 mg elacestrant hydrochloride; elacestrant 258 mg is equivalent to 300 mg elacestrant hydrochloride; and elacestrant 345 mg is equivalent to 400 mg elacestrant hydrochloride; [a]Elacestrant 258 mg + alpelisib 200 mg (cohort -1); [b]Elacestrant 258 mg* + alpelisib 150 mg (cohort -2); [c]Elacestrant 86 mg* + ribociclib (cohort 1); [d]Elacestrant 172 mg* + ribociclib (cohort 2); [e]Elacestrant 345 mg* + everolimus 7.5 mg (cohort 4); [f]Elacestrant 258 mg* + ribociclib 400 mg (cohort 3); [g]Elacestrant 172 mg* + ribociclib 600 mg (cohort 4); [h]Elacestrant 345 mg + ribociclib 400 mg (cohort 5); [i]The RP2D for the combination of elacestrant and abemaciclib was determined in the ongoing ELECTRA trial (NCT05386108). +, additional Cohorts; CDK4/6i, cyclin-dependent kinase 4/6 inhibitor; PFS, progression-free survival; RP2D, recommended phase 2 dose.

Eligibility Criteria

Key Inclusion Criteria

- Women or men age ≥18 years
- Female patients may be post-, pre-*, or perimenopausal*
- Histopathologic or cytologic confirmation of ER+, HER2- breast cancer as per the ASCO/CAP guidelines
- ≥1 measurable lesion as per RECIST v1.1 or a mainly lytic bone lesion
- ECOG PS 0–1
- **1-2 prior lines of ET, one with CDK4/6i (alpelisib,† everolimus, palbociclib, ribociclib, and abemaciclib):‡**
 - **Alpelisib combination:** *PIK3CA*-mut by local lab
 - **Capivasertib combination:§** *PIK3CA/AKT1/PTEN*-alteration as detected by an approved test (local result)
 - 1-2 prior hormonal therapies in the advanced or metastatic setting, or radiological evidence of BC recurrence or progression ≤12 months from the end of adjuvant treatment with ET. Prior CDK4/6i is allowed
- Phase 2 is the same as Phase 1b, except no prior CDK4/6i is allowed for Arm D

Key Exclusion Criteria

- Active or newly diagnosed central nervous system metastases, including meningeal carcinomatosis
- Advanced, symptomatic visceral spread with risk of life-threatening complications in the short term
- Prior chemotherapy in the advanced or metastatic setting
- Prior therapy with elacestrant or other investigational compounds in the advanced or metastatic setting
 - Prior treatment with fulvestrant is allowed
- **Alpelisib combination:** Type 1 diabetes or uncontrolled type 2 diabetes¥
- **Ribociclib combination:** QTcF values ≥450 msec; patients who are at significant risk of developing QTc prolongation;‖ currently receiving or received drugs known to prolong QT interval ≤14 days or 5 half-lives, whichever is shorter, before the first dose of trial therapy
- **Capivasertib combination:** Clinically significant abnormalities of glucose metabolism¶

*Must be concurrently receiving a LHRH agonist at least 4 weeks before the start of trial therapy and are planning to continue LHRH agonist treatment during the study treatment. †Excludes prior therapy with alpelisib or any other PI3Ki ‡Excludes prior treatment in the advanced setting with the companion drug. §Excludes prior therapy with AKTi, PI3Ki and mTORi. ¥Fasting plasma glucose level of >140 mg/dL [7.7 mmol/L], or HbA1c level >6.4%. ‖Patients with long QT syndrome; patients with uncontrolled or significant cardiac disease including recent (6 mo) myocardial infarction, congestive heart failure, unstable angina, and brady-arrhythmias; patients with electrolyte abnormalities. ¶Defined by any of the following: patients with diabetes mellitus type 1, patients with diabetes mellitus type 2 requiring insulin treatment, or patients with HbA1c level of >8.0%

RESULTS

Baseline Characteristics

	Elacestrant + Everolimus n=23	Elacestrant + Ribociclib n=33
Median age, years	64 (38-76)	66 (45-85)
Gender at birth, n (%)		
Female	23 (100)	33 (100)
Male	0	0
ECOG PS, n (%)		
0	15 (65)	24 (73)
1	8 (35)	9 (27)
Visceral metastasis,* n (%)	14 (61)	22 (67)
Mutations, n (%)		
ESR1	10 (43)	13 (39)
PIK3CA	6 (26)	10 (30)
Primary endocrine resistance,‡ n (%)	4 (17)	3 (9)
Median number of prior therapies for adv/mBC, n (range)	2 (1-3)	1 (1-2)
Prior CDK4/6i for adv/mBC, n (%)	23 (100)	33 (100)
Number of prior lines of ET for adv/mBC, n (%)		
0	1 (4)	0
1	14 (61)	26 (79)
2	8 (35)	7 (21)
Prior fulvestrant for adv/mBC, n (%)	11 (48)	18 (55)

Data cut-off: 12 MAR 2025
*Includes lung, liver, brain, pleural, and peritoneal involvement; ‡Relapse within the first two years of adjuvant on ET or progressive disease within the first six months of first-line ET for advanced breast cancer[23]
CDK4/6i, cyclin dependent kinase 4/6 inhibitor; ECOG PS, Eastern Cooperative Oncology Group performance status; ET, endocrine therapy; adv/mBC, advanced or metastatic breast cancer.

Most Common TEAEs with Elacestrant + Everolimus (RP2D)*

	Cohort 1 (n=6)		Cohort 2 (n=6)		Cohort 3 (n=4)		Cohort 4 (n=7)	
	Elacestrant 258 mg QD + Everolimus 5 mg QD		Elacestrant 345 mg QD + Everolimus 5 mg QD		Elacestrant 345 mg QD + Everolimus 10 mg QD		RP2D Elacestrant 345 mg QD + Everolimus 7.5 mg QD	
Preferred Term, n (%)	All Grades	Grade 3/4	All Grades	Grade 3/4	All Grades	Grade 3/4	All Grades	Grade 3/4
Fatigue	3 (50)	0	3 (50)	1 (17)	1 (25)	0	**4 (57)**	**0**
Diarrhea	4 (67)	0	2 (33)	1 (17)	1 (25)	1 (25)	**3 (43)**	**0**
Nausea	4 (67)	0	2 (33)	1 (17)	3 (75)	0	**3 (43)**	**0**
Vomiting	2 (33)	1 (17)	0	0	1 (25)	0	**3 (43)**	**0**
Dysgeusia	1 (17)	0	1 (17)	0	1 (25)	0	**3 (43)**	**0**
Decreased appetite	2 (33)	0	1 (17)	1 (17)	1 (25)	0	**3 (43)**	**0**
Stomatitis	3 (50)	0	2 (33)	0	1 (25)	0	**2 (29)**	**1 (14)**
Neutropenia	2 (33)	2 (33)	0	0	2 (50)	1 (25)	**2 (29)**	**1 (14)**
Insomnia	2 (33)	0	0	0	1 (25)	0	**2 (29)**	**0**
Pain in extremity	0	0	1 (17)	0	0	0	**2 (29)**	**0**

Data cut-off: 12 MAR 2025. *TEAE ≥15% at RP2D.
RP2D, Recommended phase 2 dose; TEAEs, Treatment-emergent adverse events.

Most Common TEAEs with Elacestrant + Ribociclib (RP2D)*

	Cohort 1 (n=6)		Cohort 2 (n=6)		Cohort 3 (n=6)		Cohort 4 (n=7)		Cohort 5 (n=8)	
	Elacestrant 86 mg QD + Ribociclib 400 mg QD		Elacestrant 172 mg QD + Ribociclib 400 mg QD		Elacestrant 258 mg QD + Ribociclib 400 mg QD		Elacestrant 172 mg QD + Ribociclib 600 mg QD		RP2D Elacestrant 345 mg QD + Ribociclib 400 mg QD	
Preferred Term, n (%)	All Grades	Grade 3/4	All Grades	Grade 3/4	All Grades	Grade 3/4	All Grades	Grade 3/4	All Grades	Grade 3/4
Neutropenia	3 (50)	2 (33)	4 (67)	2 (33)	1 (17)	1 (17)	4 (57)	4 (57)	**3 (38)**	**2 (25)**
Nausea	2 (33)	0	3 (50)	0	1 (17)	0	4 (57)	0	**2 (25)**	**0**
Fatigue	2 (33)	0	2 (33)	0	1 (17)	0	4 (57)	0	**2 (25)**	**0**
Upper respiratory tract infection	1 (17)	0	0	0	0	0	0	0	**2 (25)**	**0**
White blood cell count decrease	0	0	0	0	1 (17)	0	3 (43)	2 (29)	**2 (25)**	**1 (13)**

Data cut-off: 12 MAR 2025. *TEAE ≥15% at RP2D.
RP2D, Recommended phase 2 dose; TEAEs, Treatment-emergent adverse events.

Response and Clinical Benefit in Phase 1b Efficacy-Evaluable Patients

Efficacy Outcome* (mRECIST)	Elacestrant + Everolimus n=16	Elacestrant + Ribociclib n=31
ORR, n (%)	4 (25)	3 (10)
CR	-	-
PR	4 (25)	3 (10)
SD	7 (44)	22 (71)
PD	5 (31)	6 (19)
CBR, n (%)	11 (69)	25 (81)

Data cut-off: 12 MAR 2025.
*Confirmed responses only; includes patients who had measurable disease (ie, at least 1 target lesion) at baseline and at least 1 post-baseline assessment using Modified Response Evaluation Criteria in Solid Tumors (mRECIST).

Elacestrant + Everolimus: Phase 1b Best Tumor Response*

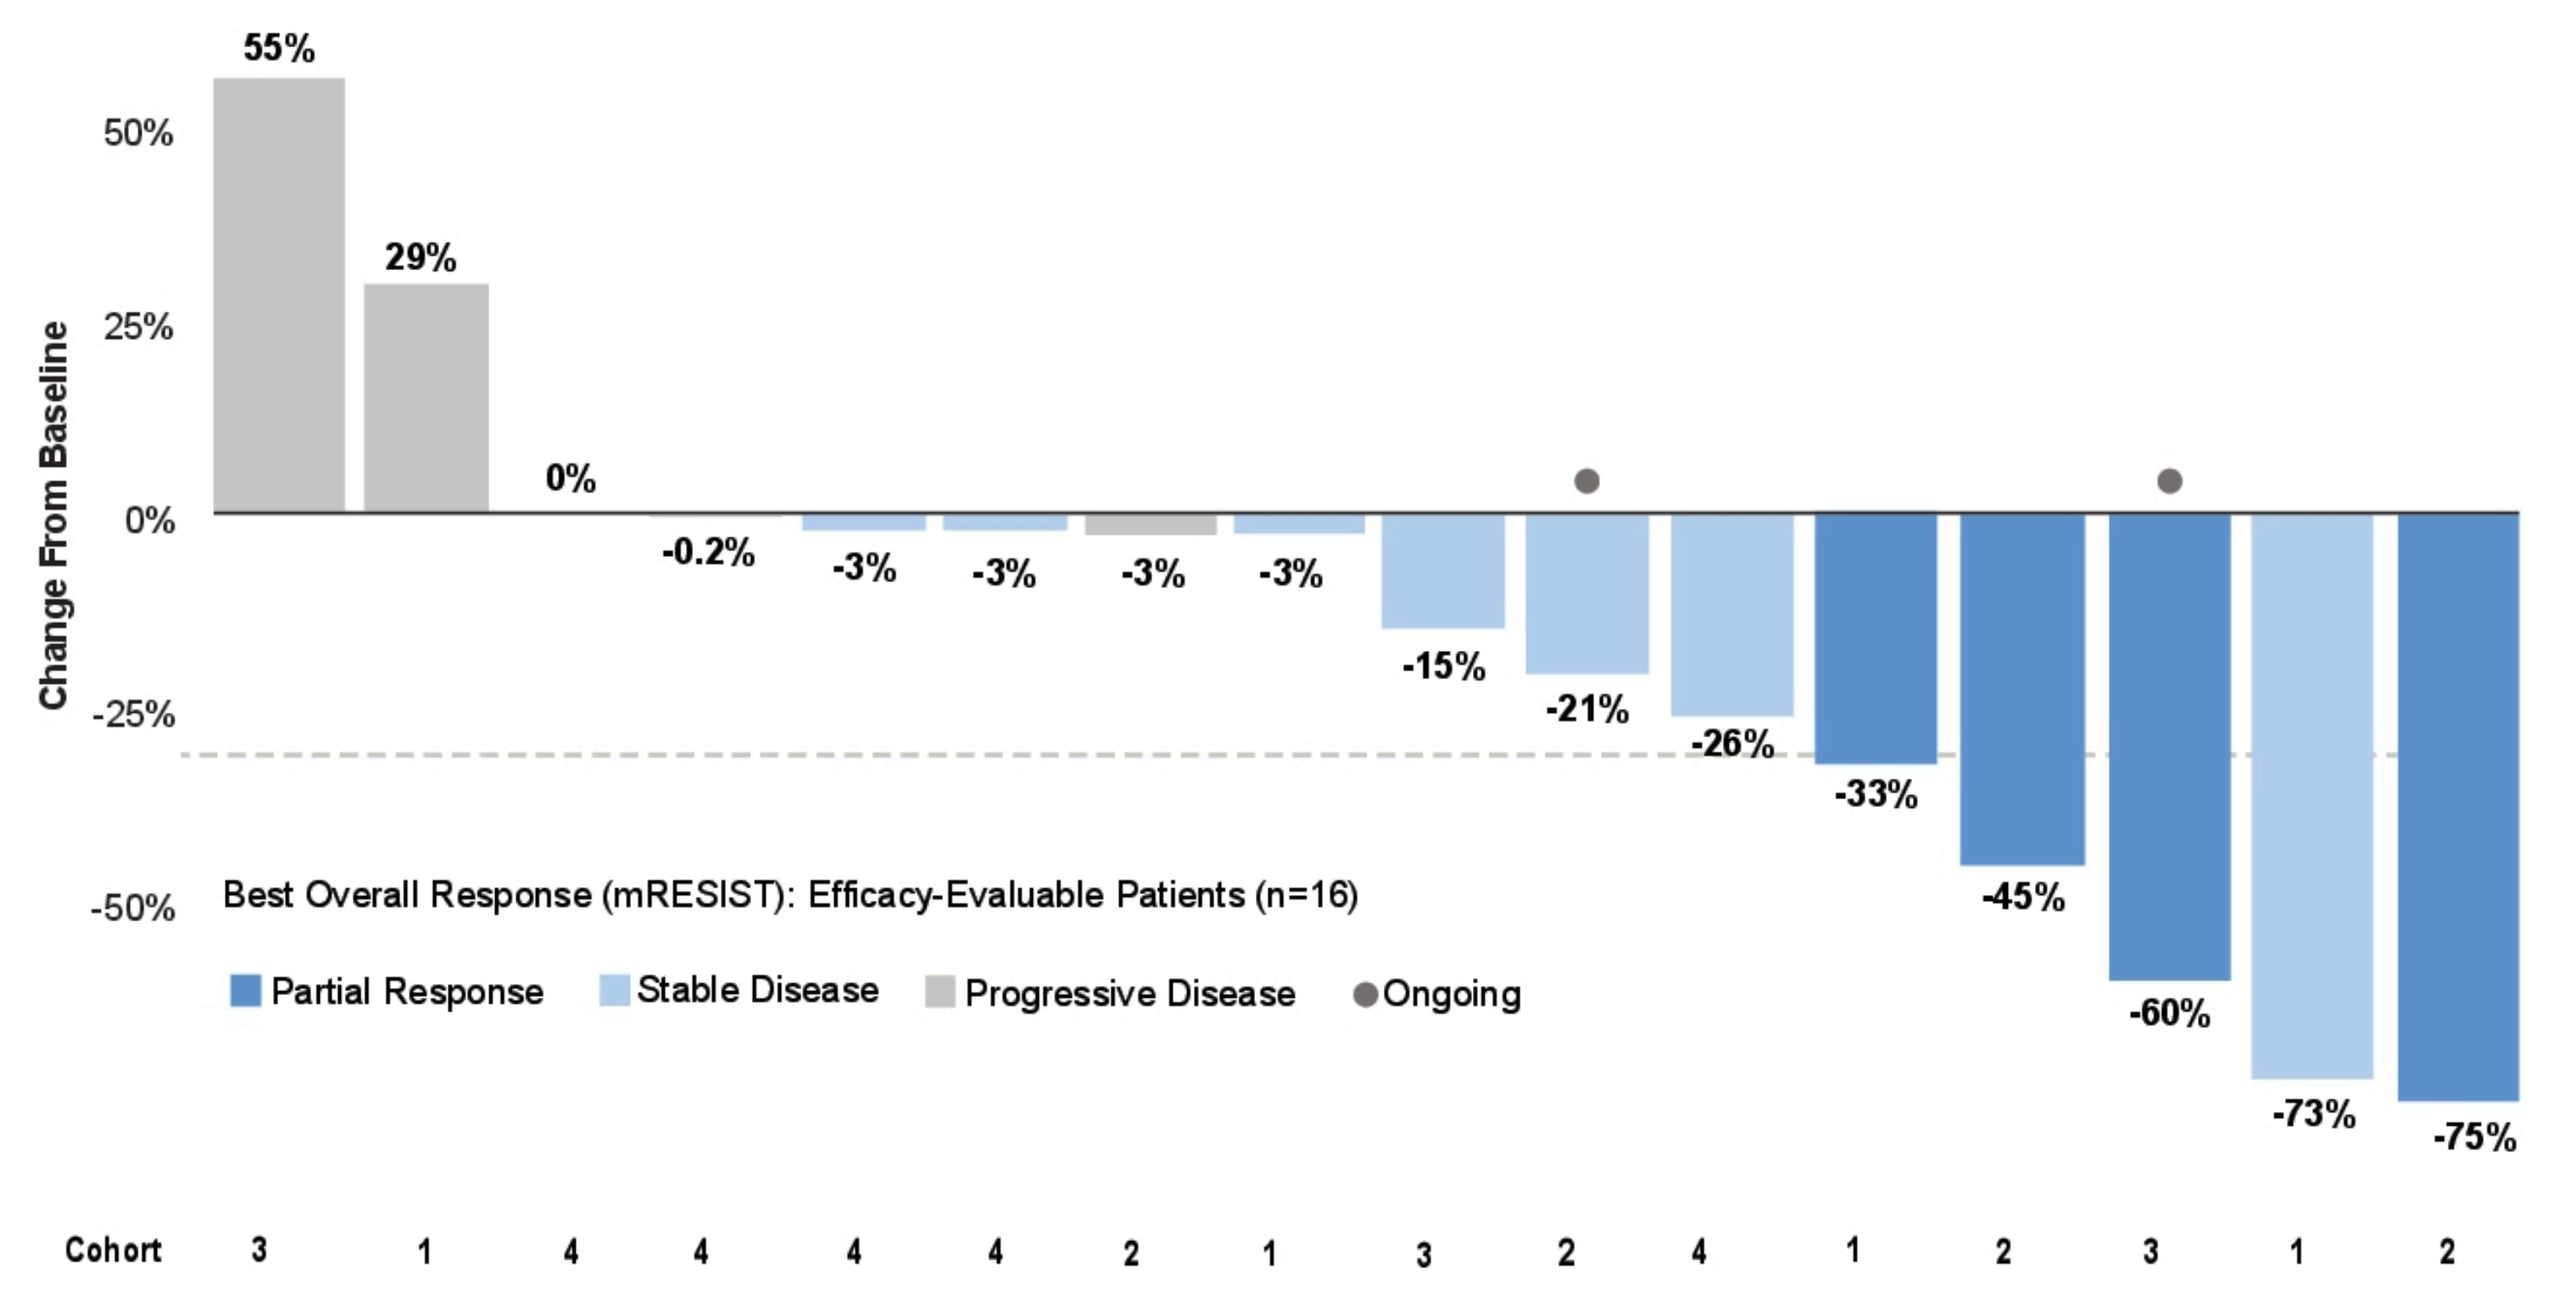
Change From Baseline
50%
25%
0%
-25%
-50%
55%
29%
0%
-0.2%
-3%
-3%
-3%
-3%
-15%
-21%
-26%
-33%
-45%
-60%
-73%
-75%
Best Overall Response (mRESIST): Efficacy-Evaluable Patients (n=16)
Partial Response
Stable Disease
Progressive Disease
Ongoing
Cohort 3 1 4 4 4 4 2 1 3 2 4 1 2 3 1 2

Data cut-off: 12 MAR 2025
*Confirmed responses only; includes patients who had measurable disease (ie, at least 1 target lesion) at baseline and at least 1 post-baseline assessment using Modified Response Evaluation Criteria in Solid Tumors (mRECIST).

Elacestrant + Ribociclib: Phase 1b Best Tumor Response*

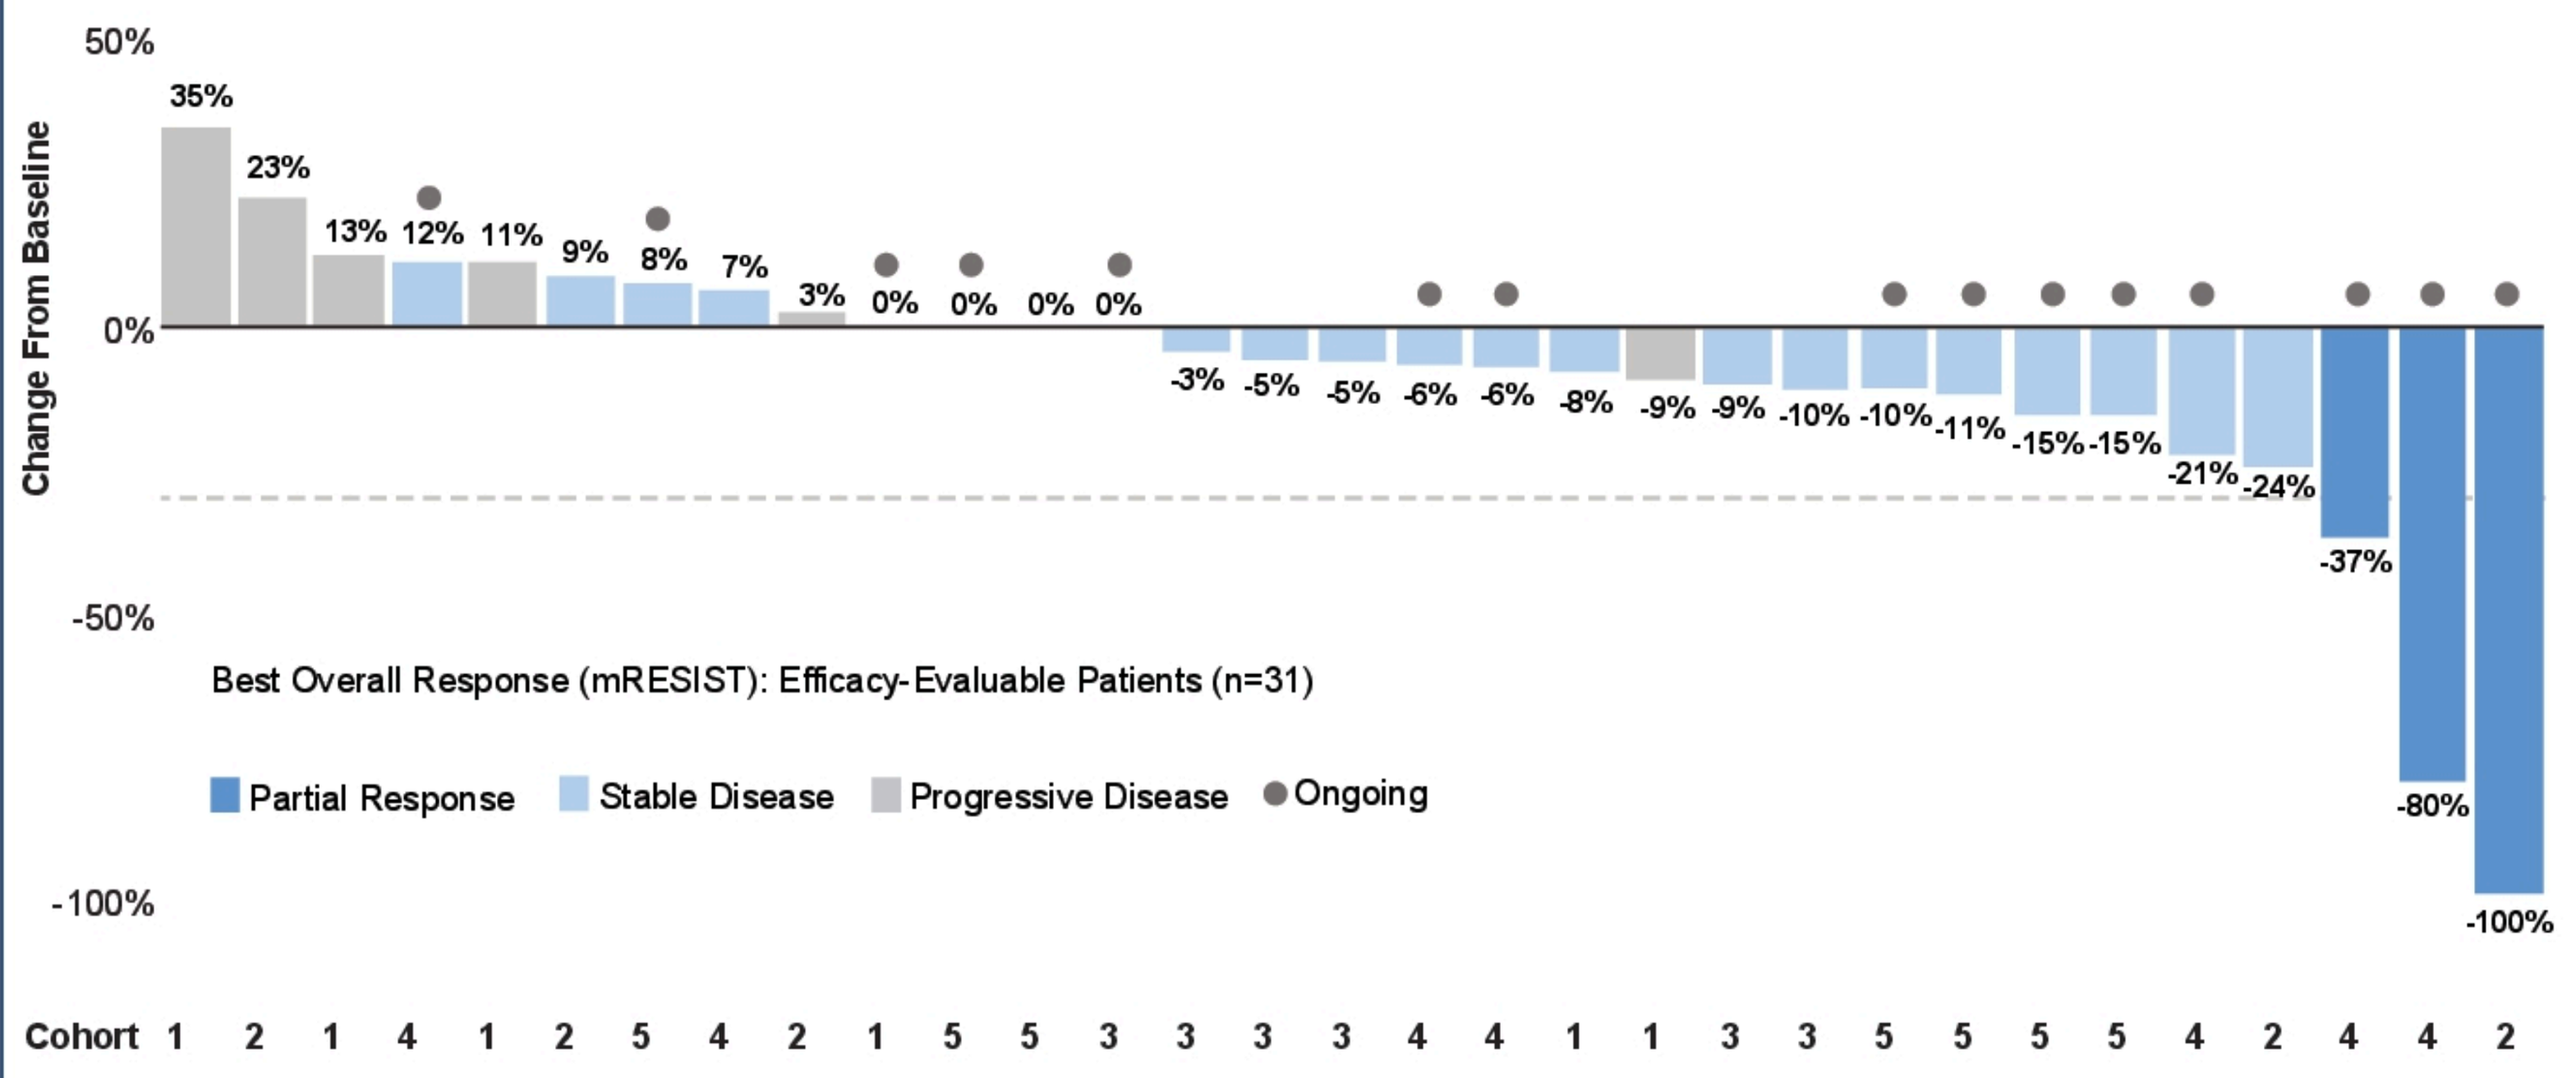
Change From Baseline
50%
0%
-50%
-100%
35%
23%
13%
12%
11%
9%
8%
7%
3%
0%
0%
0%
0%
-3%
-5%
-5%
-6%
-6%
-8%
-9%
-9%
-10%
-10%
-11%
-15%
-15%
-21%
-24%
-37%
-80%
-100%
Best Overall Response (mRESIST): Efficacy-Evaluable Patients (n=31)
Partial Response
Stable Disease
Progressive Disease
Ongoing
Cohort 1 2 1 4 1 2 5 4 2 1 5 5 3 3 3 3 4 4 1 1 3 3 5 5 5 4 2 4 4 2

Data cut-off: 12 MAR 2025
*Confirmed responses only; includes patients who had measurable disease (ie, at least 1 target lesion) at baseline and at least 1 post-baseline assessment using Modified Response Evaluation Criteria in Solid Tumors (mRECIST).

mPFS in Phase 1b All Patients

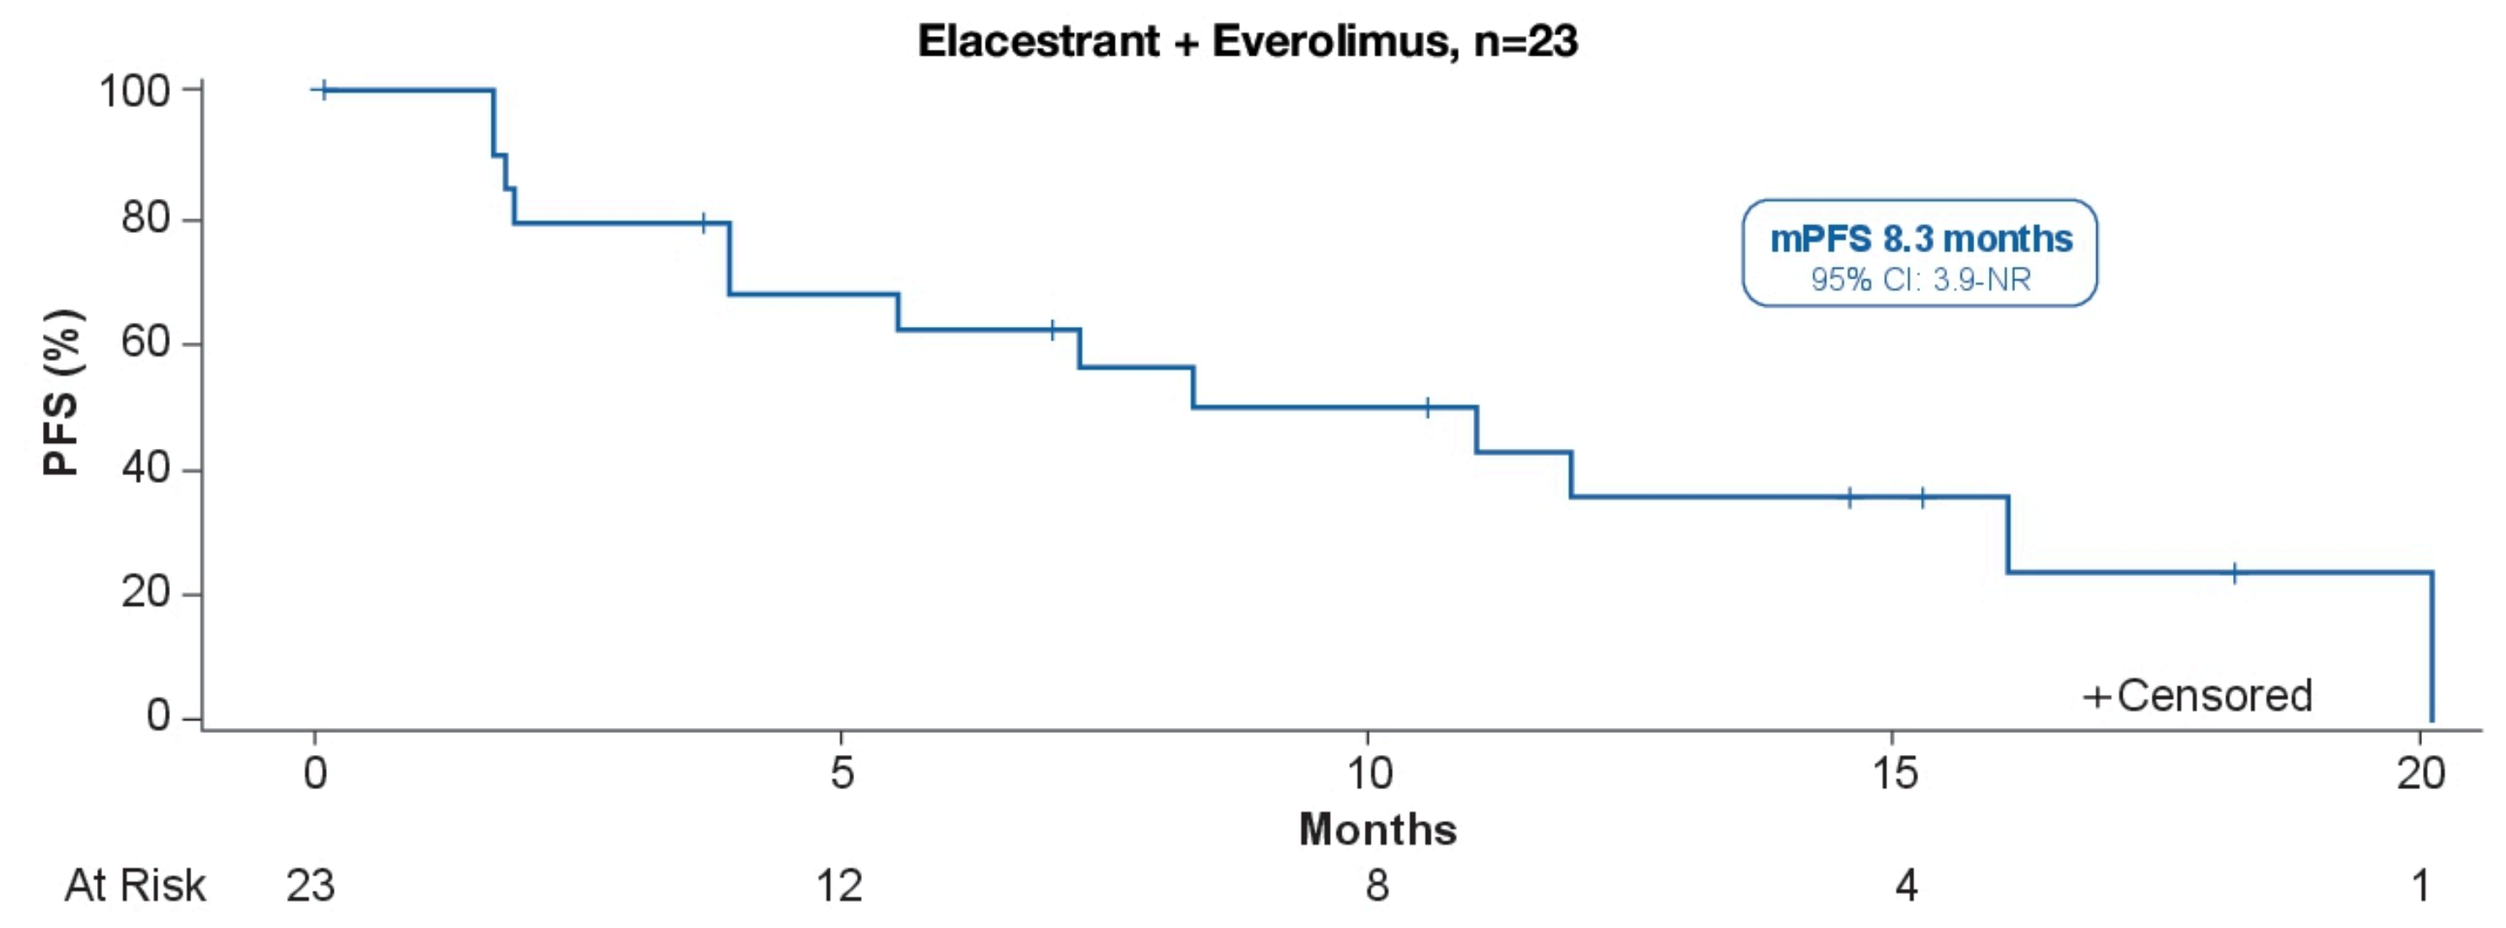
Elacestrant + Everolimus, n=23
PFS (%)
100
80
60
40
20
0
mPFS 8.3 months
95% CI: 3.9-NR
+Censored
0 5 10 15 20
Months
At Risk 23 12 8 4 1

Median follow-up time of 5.5 months at data cut-off

- Clinical characteristics: visceral metastases 61%, prior fulvestrant 48%, 2 prior lines of ET for adv/mBC 35%, primary endocrine resistance 17%, *ESR1*-mutated tumors 43% (10/23), and *PIK3CA*-mutated tumors 26% (6/23).

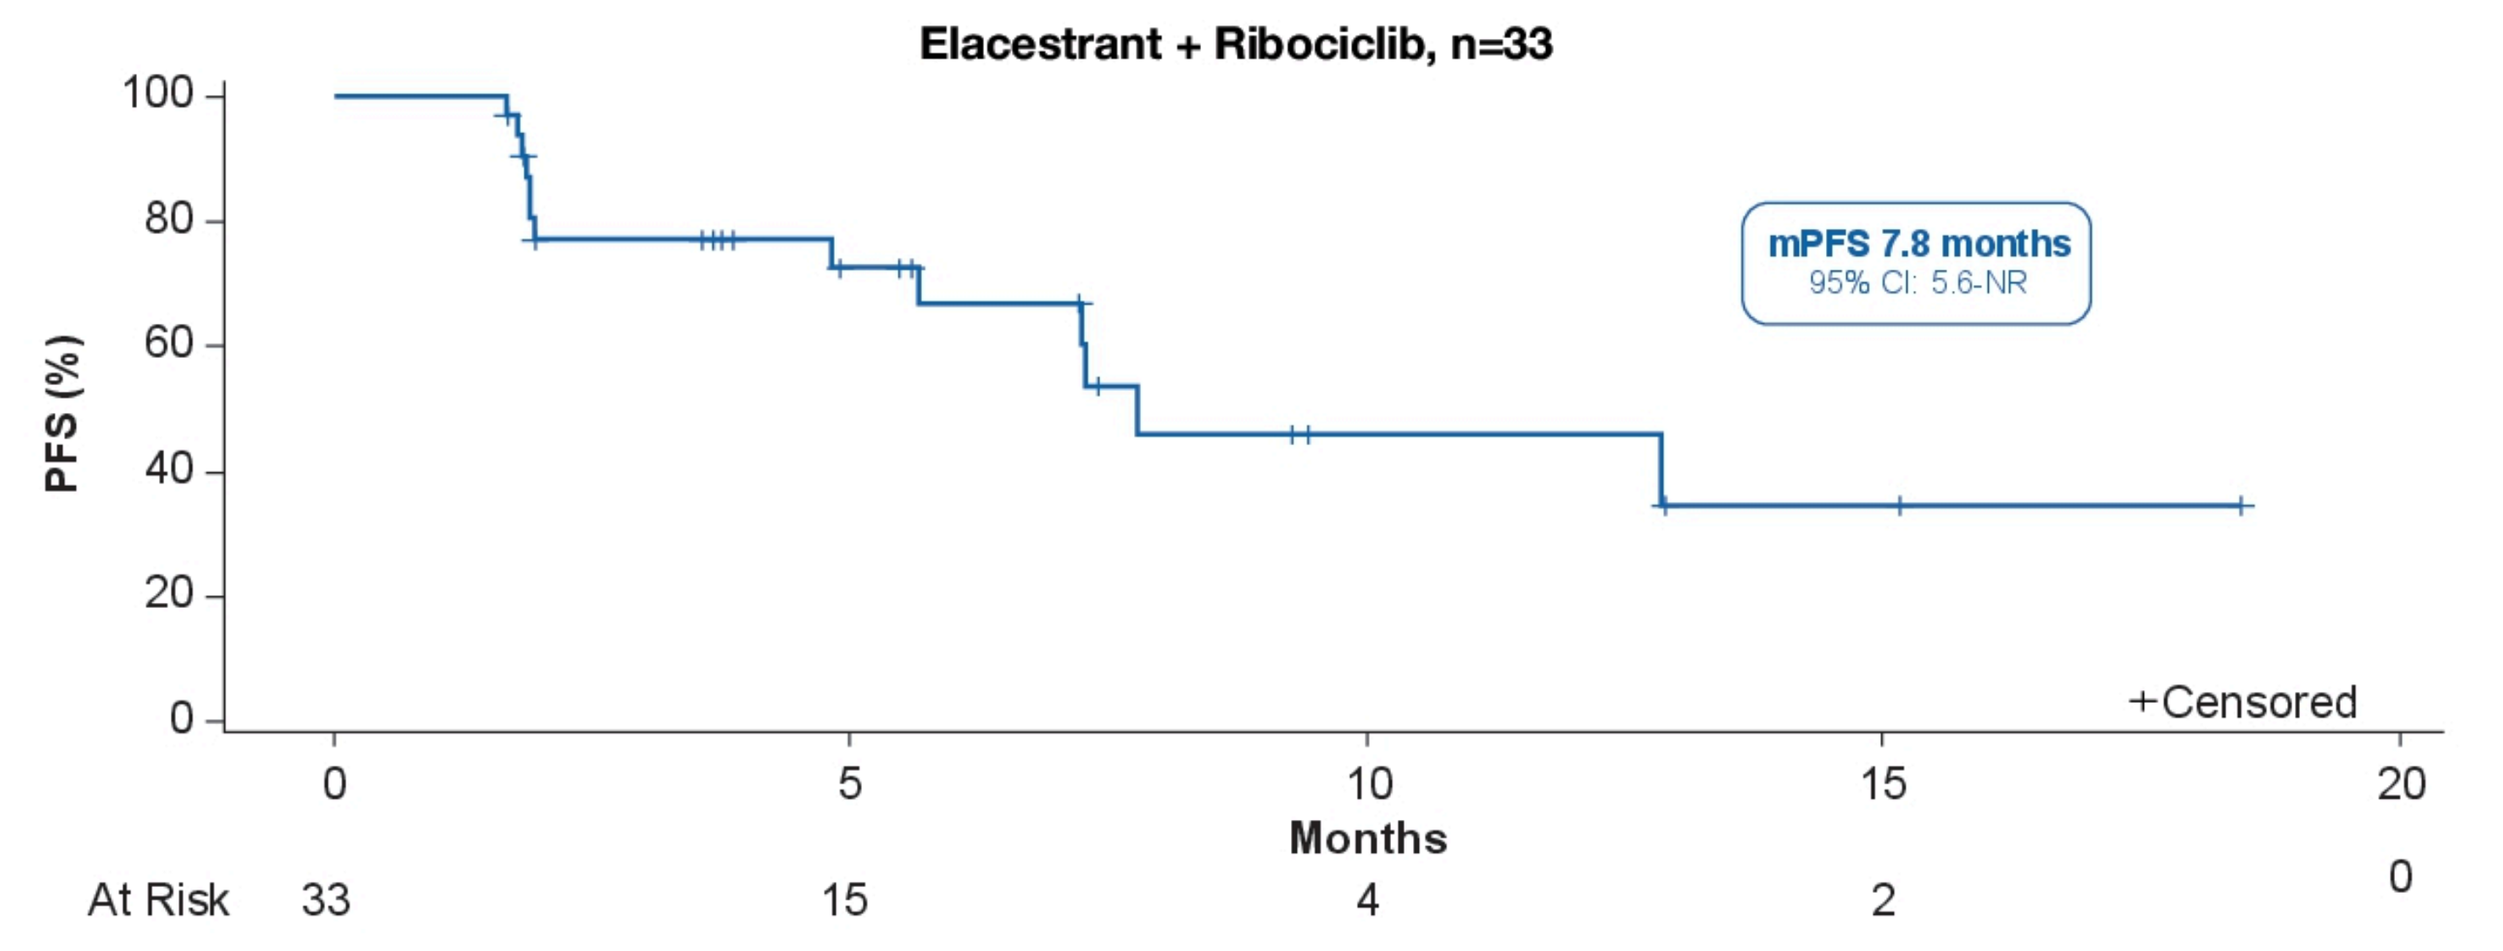
Elacestrant + Ribociclib, n=33
PFS (%)
100
80
60
40
20
0
mPFS 7.8 months
95% CI: 5.6-NR
+Censored
0 5 10 15 20
Months
At Risk 33 15 4 2 0

Median follow-up time of 4.8 months at data cut-off

- Clinical characteristics: visceral metastases 67%, prior fulvestrant 55%, 2 prior lines of ET for adv/mBC 21%, primary endocrine resistance 9%, *ESR1*-mutated tumors 39% (13/33), and *PIK3CA*-mutated tumors 30% (10/33).

Data cut-off: 12 MAR 2025.

CONCLUSIONS

- In patients with prior ET + CDK4/6i, the safety of elacestrant + everolimus and elacestrant + ribociclib combinations were consistent with the known profile of everolimus or ribociclib combined with standard ET, and no new signals were observed.
- The phase 1b combinations evaluated showed clinically important efficacy regardless of *ESR1*-mutation status.
 - mPFS was 8.3 months with elacestrant + everolimus in all patients.
 - mPFS was 7.8 months with elacestrant + ribociclib in all patients.
- The following phase 2 combinations are ongoing.
 - Elacestrant 345 mg + everolimus 7.5 mg: ELEVATE (Arm B)
 - Elacestrant 345 mg + ribociclib 400 mg: ELEVATE (Arm C)
 - Elacestrant 345 mg + abemaciclib 150 mg BID: ELEVATE (Arms C and D)
- Phase 1b combinations for alpelisib and capivasertib are ongoing.
- Elacestrant has the potential to become an ET backbone for multiple targeted agents in patients with ER+/HER2- mBC due to its ability to extend PFS, provide an all-oral treatment option, and delay chemotherapy or ADC-based regimens.

REFERENCES:
1. Osborne CK, et al. Annu Rev Med. 2011;62:233-247; 2. Brett JO, et al. Breast Cancer Res. 2021;23(1):85; 3. Bidard FC, et al. Lancet Oncol. 2022;23(11):1367-1377; 4. Santiago Novello RG, et al. ESMO Open. 2023; 8(suppl 4):104409; 5. Glaviano A, et al. Mol Cancer. 2023;22(1):138; 6. Cancer Genome Atlas Network. Nature. 2012;490(7418):61-70; 7. Hartkopf AD, et al. Breast Care (Basel). 2020;15:347-54; 8. Lei JT, et al. Breast. 2019;48:S26–S30; 9. Jeselsohn R, et al. Clin Cancer Res. 2014;20(7):1757-1767; 10. Toy W, et al. Nat Genet. 2013;45(12):1439-1445; 11. Oesterreich S, et al. Nat Genet. 2013;45(12):1415-1416; 12. Lloyd MR, et al. Ther Adv Med Oncol. 2022;14:17588359221113694; 13. Robinson DR, et al. Nat Gen. 2013;45:1446-51; 14. Jhaveri K, et al. Ann Oncol. 2023;34(suppl_2): S334-S390; 15. Bhave MA, et al. SABCS 2023. PO2-16-05. 16. Cook MM, et al. Oncologist. 2021;26(2):101-106; 17. Vasseur A, et al. Oncogene. 2024;43(16):1214-1222; 18. Kalinsky K, et al. J Clin Oncol. 2023;41(24):4004-4013; 19.Bidard FC, et al. J Clin Oncol. 2022;40:3246-3256; 20. Bardia A, et al. Clin Cancer Res. 2024;30(19):4299-4309; 21. Bihani T, et al. Clin Cancer Res. 2017;23:4793-4804; 22. Patel HK, et al. Breast Cancer Res. 2019;21:146; 23. Cardoso F, et al. Breast. 2024;76:103756

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® and the author of this poster