Elacestrant Versus Fulvestrant or Aromatase Inhibitor in a Phase 3 Trial Evaluating Elacestrant, an Oral Selective Estrogen Receptor Degrader Versus Standard of Care Endocrine Monotherapy for ER+/HER2- Advanced/Metastatic Breast Cancer: Subgroup Analysis from EMERALD

Presented at ESMO 2022

Presentation: 220P

Aftimos P,[1] Cortes J,[2] Bidard FC,[3] Kaklamani V,[4] Bardia A,[5] Neven P,[6] Streich G,[7] Montero AJ,[8] Forget F,[9] Mouret-Reynier MA,[10] Sohn JH,[11] Taylor D,[12] Harnden KK,[13] Khong H,[14] Kocsis J,[15] Dalenc F,[16] Dillon P,[17] Tonini G,[18] Grzegorzewski KJ,[19] Lu J[20]

[1]Institut Jules Bordet – Université Libre de Bruxelles, Brussels, Belgium; [2]International Breast Cancer Center (IBCC), Quiron Group, Barcelona, Spain; [3]Institut Curie, Paris and Saint-Cloud, France; [4]University of Texas Health Sciences Center, Houston, TX, USA; [5]Massachusetts General Hospital Cancer Center, Harvard Medical School, Boston, MA, USA; [6]Universitaire Ziekenhuizen (UZ) – Leuven Cancer Institute, Leuven, Belgium; [7]Centro Médico Austral, Buenos Aires, Argentina; [8]University Hospitals Seidman Cancer Center – Case Western Reserve University, Cleveland, OH, USA; [9]Centre Hospitalier de l'Ardenne – Site de Libramont, Libramont-Chevigny, Belgium; [10]Centre Jean Perrin, Clermont-Ferrand, France; [11]Yonsei Cancer Center, Yonsei University Health System – Medical Oncology, Seoul, Republic of Korea; [12]Université Catholique de Louvain, CHU UCL Namur – Site Sainte-Elisabeth, Namur, Belgium; [13]Inova Schar Cancer Institute, Fairfax, VA, USA; [14]Moffit Cancer Center & Research Institute, Tampa, FL, USA; [15]Bács-Kiskun Megyei Kórház, Kecskemét, Hungary; [16]Institut Claudius Regaud, IUCT-Oncopole, Toulouse, France; [17]University of Virginia Cancer Center, Charlottesville, VA, USA; [18]Menarini Group, Florence, Italy; [19]Stemline Therapeutics/Menarini Group, New York, NY, USA; [20]University Of Southern California/Norris Comprehensive Cancer Center, Los Angeles, CA, USA

BACKGROUND

- Elacestrant is a novel, oral selective estrogen receptor degrader (SERD).
- The phase 3 EMERALD trial compared the efficacy and safety of elacestrant to standard-of-care (SOC) endocrine therapy of investigator's choice (fulvestrant or aromatase inhibitors [AI]) in patients with ER+/HER2- locally advanced or metastatic breast cancer (mBC) following progression on prior endocrine and cyclin-dependent kinase 4/6 inhibitor (CDK4/6i) therapy.[1]
- Elacestrant demonstrated significantly prolonged progression-free survival (PFS) compared to SOC in all patients and in patients whose tumors harbored *ESR1* mutations (*ESR1*-mut).[1]
- Here, we report a post-hoc subgroup analysis from EMERALD separately comparing the efficacy of elacestrant to fulvestrant and to AI.

METHODS

- EMERALD (NCT03778931) is a randomized, open-label, phase 3 trial (**Figure 1**).[1]
- Patients were randomized 1:1 to elacestrant (400 mg orally daily) or SOC consisting of investigator's choice of fulvestrant or AI.
- For the SOC arm, the protocol provided the following guidance regarding therapy selection:
 - Patients who have not previously received fulvestrant should be treated with fulvestrant.
 - Patients who have progressed on prior fulvestrant therapy should be treated with an AI.
 - The selection of an AI should be based on prior AI therapy and any known contraindications.
 - If the patient has previously progressed on a non-steroidal AI (anastrozole or letrozole) but not received exemestane, the preferred option would be exemestane.
 - If the patient has previously progressed on exemestane but not received a non-steroidal AI, the preferred option would be a non-steroidal AI.

Figure 1. EMERALD Phase 3 Study Design

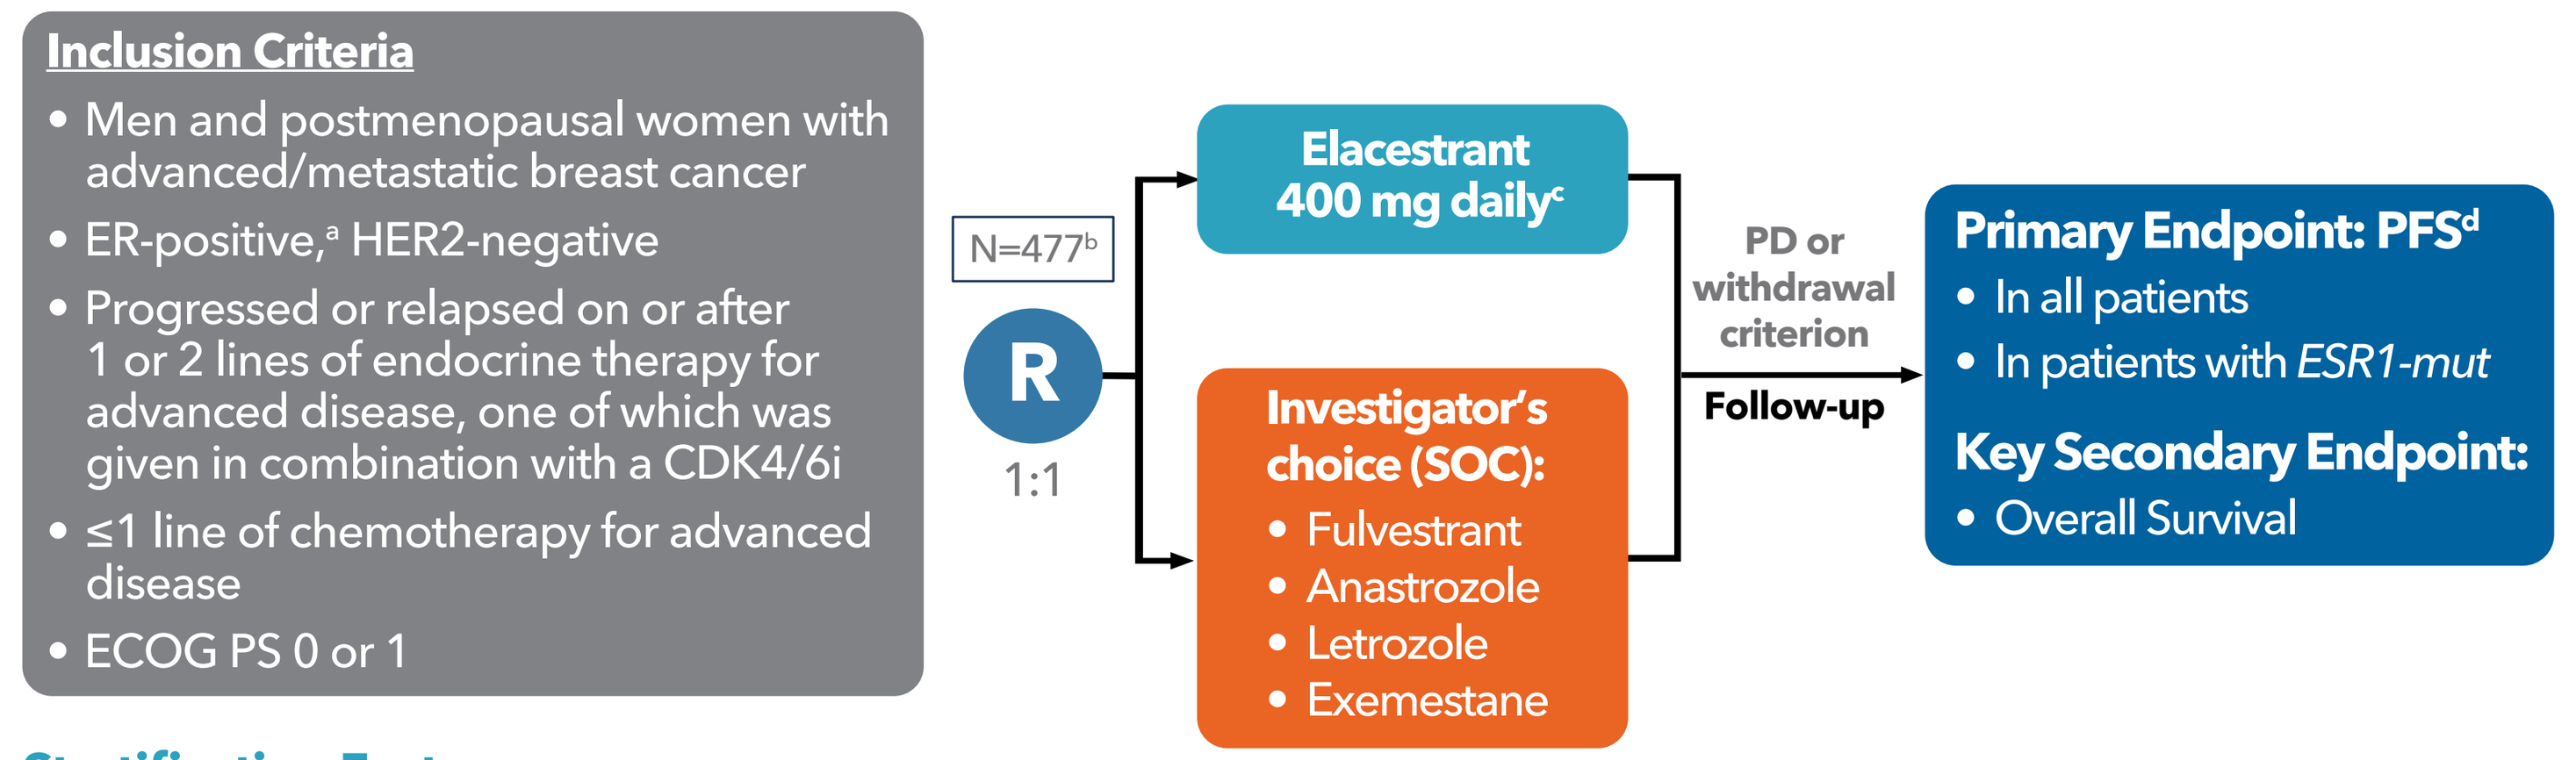
Inclusion Criteria
• Men and postmenopausal women with advanced/metastatic breast cancer
• ER-positive,a HER2-negative
• Progressed or relapsed on or after 1 or 2 lines of endocrine therapy for advanced disease, one of which was given in combination with a CDK4/6i
• ≤1 line of chemotherapy for advanced disease
• ECOG PS 0 or 1
N=477b
R
1:1
Elacestrant 400 mg dailyc
Investigator's choice (SOC):
• Fulvestrant
• Anastrozole
• Letrozole
• Exemestane
PD or withdrawal criterion
Follow-up
Primary Endpoint: PFSd
• In all patients
• In patients with ESR1-mut
Key Secondary Endpoint:
• Overall Survival

Stratification Factors:
- *ESR1-mut* status[e]
- Prior treatment with fulvestrant
- Presence of visceral metastases

[a]Documentation of ER+ tumor with ≥ 1% staining by immunohistochemistry; [b]Recruitment from February 2019 to October 2020; [c]Protocol-defined dose reductions permitted; [d]Blinded Independent Central Review. [e]*ESR1-mut* status was determined by ctDNA analysis using the Guardant360 assay (Guardant health, Redwood City, CA)

CBR, clinical benefit rate; CDK4/6i, cyclin-dependent kinase 4/6 inhibitor; DOR, duration of response; ECOG PS, Eastern Cooperative Oncology Group performance status; *ESR1-mut, ESR1* mutation; ORR, objective response rate; OS, overall survival, PD, progressive disease; PFS: progression-free survival; R, randomized. SOC, standard of care.

Elacestrant improved PFS compared with fulvestrant or AI in both the overall population and patients with *ESR1-mut*

ENDPOINTS

- The primary endpoint of EMERALD was PFS in all patients and in patients with *ESR1-mut*.
- This post-hoc analysis compared PFS separately between elacestrant and fulvestrant, as well as elacestrant and AI. It should be noted that the powering of the study was for SOC, and not for any subgroup analysis.

RESULTS

- Of 477 patients enrolled in the trial:
 - n=239 received elacestrant, n=238 received SOC
 - n=165 (69%) received fulvestrant (159 patients were pretreated with AI)
 - n=73 (31%) received AI (69 patients were pretreated with fulvestrant)
- Baseline characteristics were balanced between the elacestrant and SOC groups (**Table 1**).
- CDK4/6i are frequently combined with AI in the 1st-line ER+/HER2- mBC setting; therefore, patient disposition showing more fulvestrant (69%) vs AI (31%) in EMERALD reflects real-life setting and that the therapy selection guidance in the protocol was followed by the sites.
- A greater proportion of AI-treated patients had received two prior endocrine therapies (71.2%) vs fulvestrant-treated patients (27.3%).

Table 1. Baseline Characteristics[1]

Parameter	Elacestrant		SOC					
			Total		Fulvestrant		AI	
	All (N=239)	*ESR1-mut* (N=115)	All (N=238)	*ESR1-mut* (N=113)	All (N=165)	*ESR1-mut* (N=83)	All (N=73)	*ESR1-mut* (N=30)
Median age, yr (range)	63 (24-89)	64 (28-89)	64 (32-83)	63 (32-83)	63 (32-83)	62 (32-83)	67 (44-83)	68 (44-83)
Female, n (%)	233 (97.5)	115 (100)	237 (99.6)	113 (100)	164 (99.4)	83 (100)	73 (100)	30 (100)
Visceral metastasis,[a] n (%)	163 (68.2)	81 (70.4)	169 (71)	84 (74.3)	117 (70.9)	60 (72.3)	52 (71.2)	24 (80.0)
Prior CDK4/6i, n (%)	239 (100)	115 (100)	238 (100)	113 (100)	165 (100)	83 (100)	73 (100)	30 (100)
Two prior lines of endocrine therapy,[b] n (%)	110 (46.0)	42 (36.5)	97 (40.8)	44 (38.9)	45 (27.3)	19 (22.9)	52 (71.2)	25 (83.3)
Prior chemotherapy,[b,c] n (%)	48 (20.1)	26 (22.6)	58 (24.4)	32 (28.3)	33 (20.0)	19 (22.9)	25 (34.2)	13 (43.3)
Type of prior endocrine therapy,[b] n (%)								
Fulvestrant	70 (29.3)	27 (23.5)	75 (31.5)	28 (24.8)	6 (3.6)	1 (1.2)	69 (95.4)	27 (90.0)
Aromatase Inhibitors	193 (80.8)	101 (87.8)	193 (81.1)	96 (85.0)	159 (96.4)	78 (94.0)	34 (46.6)	18 (60.0)
Tamoxifen	19 (7.9)	9 (7.8)	15 (6.3)	9 (8.0)	10 (6.1)	6 (7.2)	5 (6.8)	3 (10.0)

[a]Includes lung, liver, brain, pleural, and peritoneal involvement; [b]Received in the advanced or metastatic setting; [c]One prior line of chemotherapy was permitted.

Efficacy

- PFS was improved in the elacestrant arm relative to patients in the SOC arm who received fulvestrant as well as those who received AI.
- PFS results vs fulvestrant and vs AIs are consistent with the overall PFS results, both in all patients and in patients with *ESR1-mut*.
- Protocol-specified landmark analysis at 6, 12, and 18 months showed a consistent benefit in terms of PFS estimates in favor of elacestrant relative to both fulvestrant and AI, separately, both in all patients and in patients with *ESR1-mut* (**Figure 2**).

Figure 2. Progression-Free Survival

Elacestrant (N=239) vs Fulvestrant (N=165): All Patients

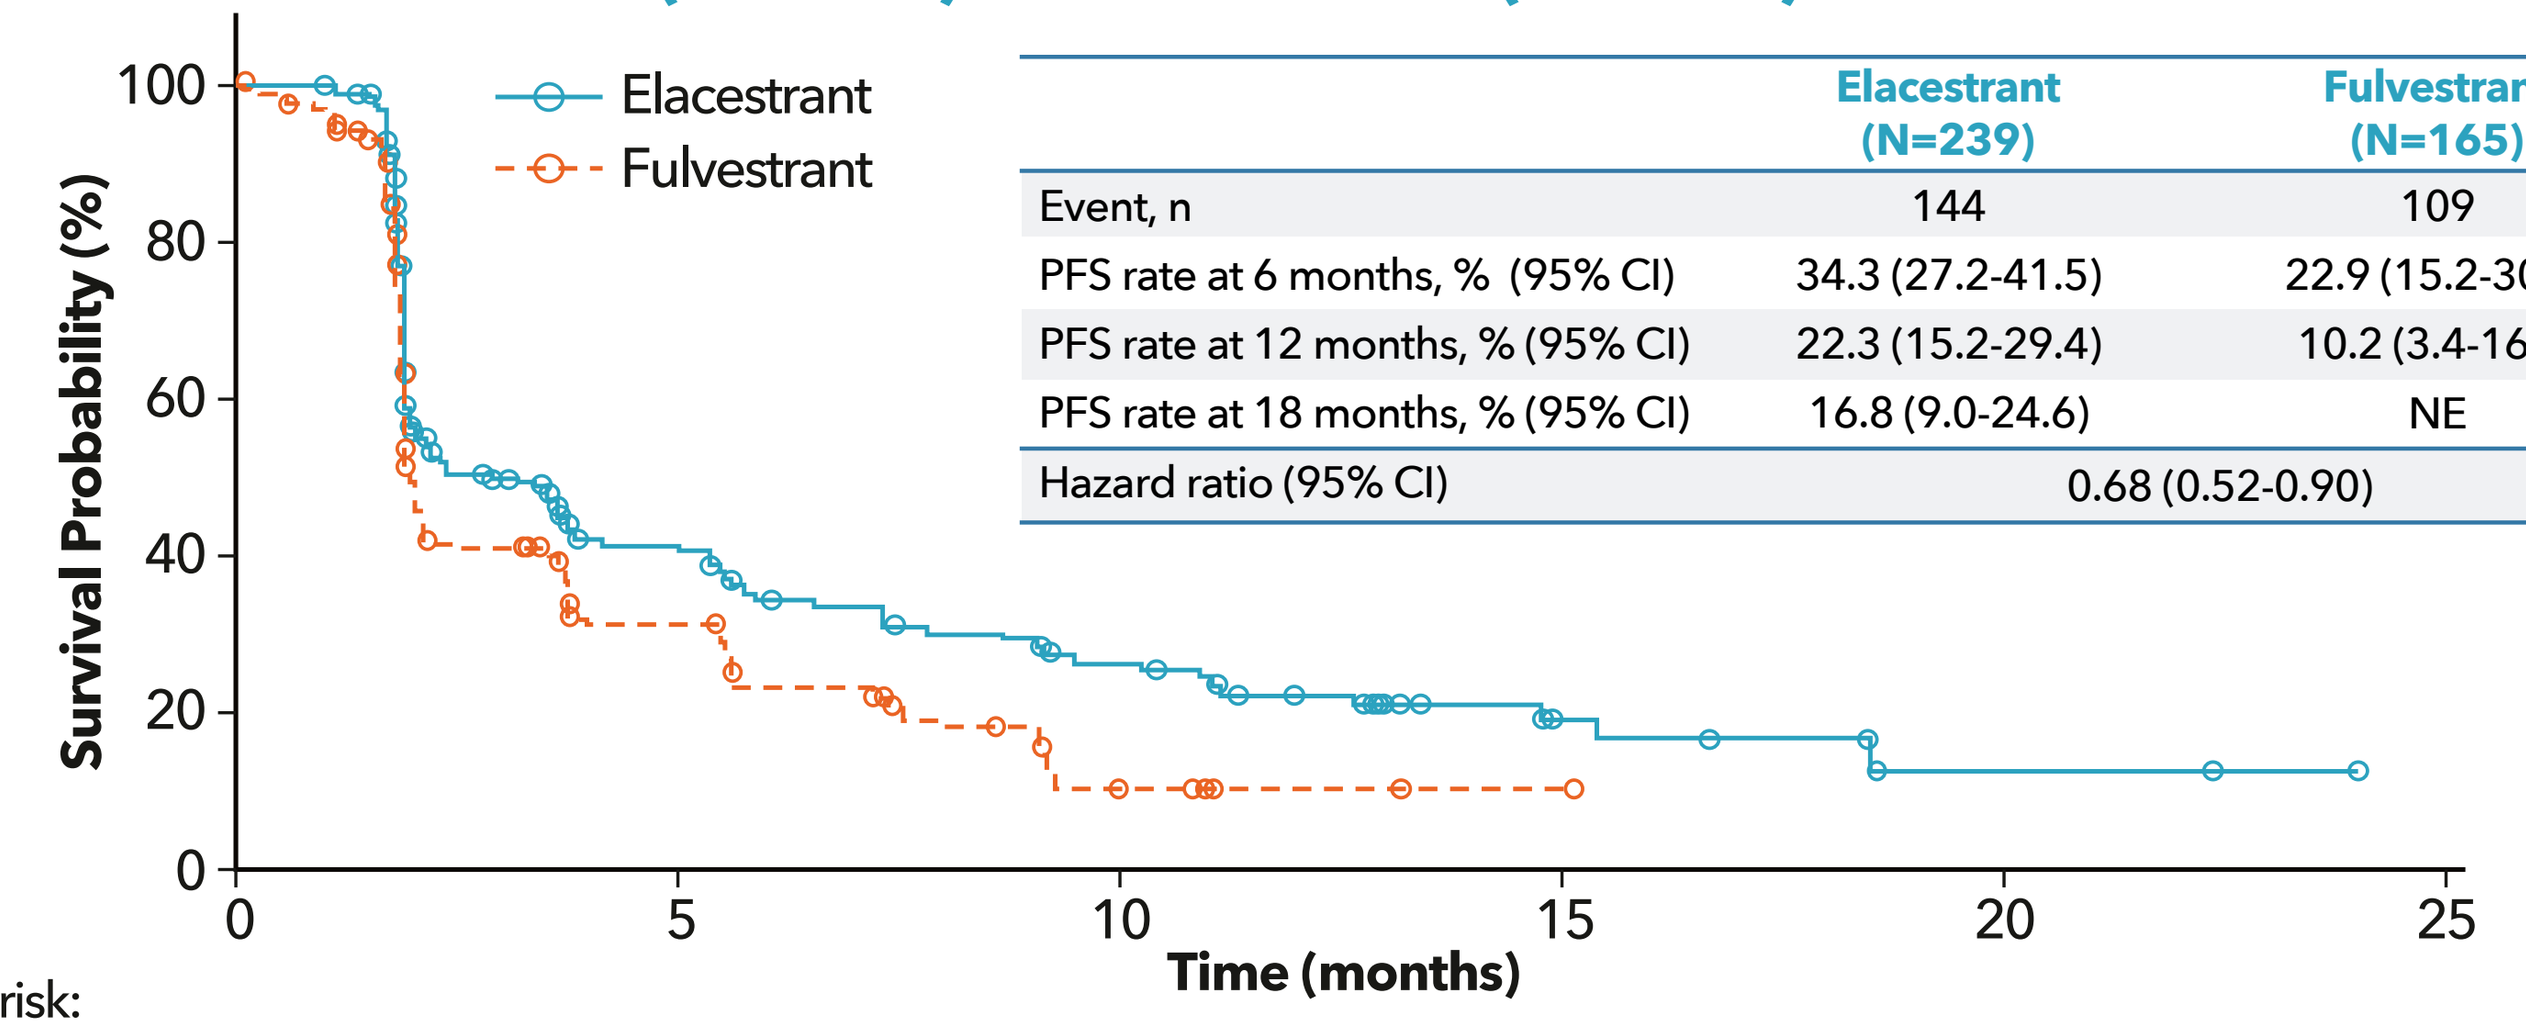
Elacestrant
Fulvestrant
Survival Probability (%)
100
80
60
40
20
0
0
5
10
15
20
25
Time (months)

	Elacestrant (N=239)	Fulvestrant (N=165)
Event, n	144	109
PFS rate at 6 months, % (95% CI)	34.3 (27.2-41.5)	22.9 (15.2-30.6)
PFS rate at 12 months, % (95% CI)	22.3 (15.2-29.4)	10.2 (3.4-16.9)
PFS rate at 18 months, % (95% CI)	16.8 (9.0-24.6)	NE
Hazard ratio (95% CI)	0.68 (0.52-0.90)	

No. at risk:											
Elacestrant	239	92	57	35	27	19	8	6	2	1	0
Fulvestrant	165	52	33	16	5	2	1	0			

Elacestrant (N=239) vs AI (N=73): All Patients

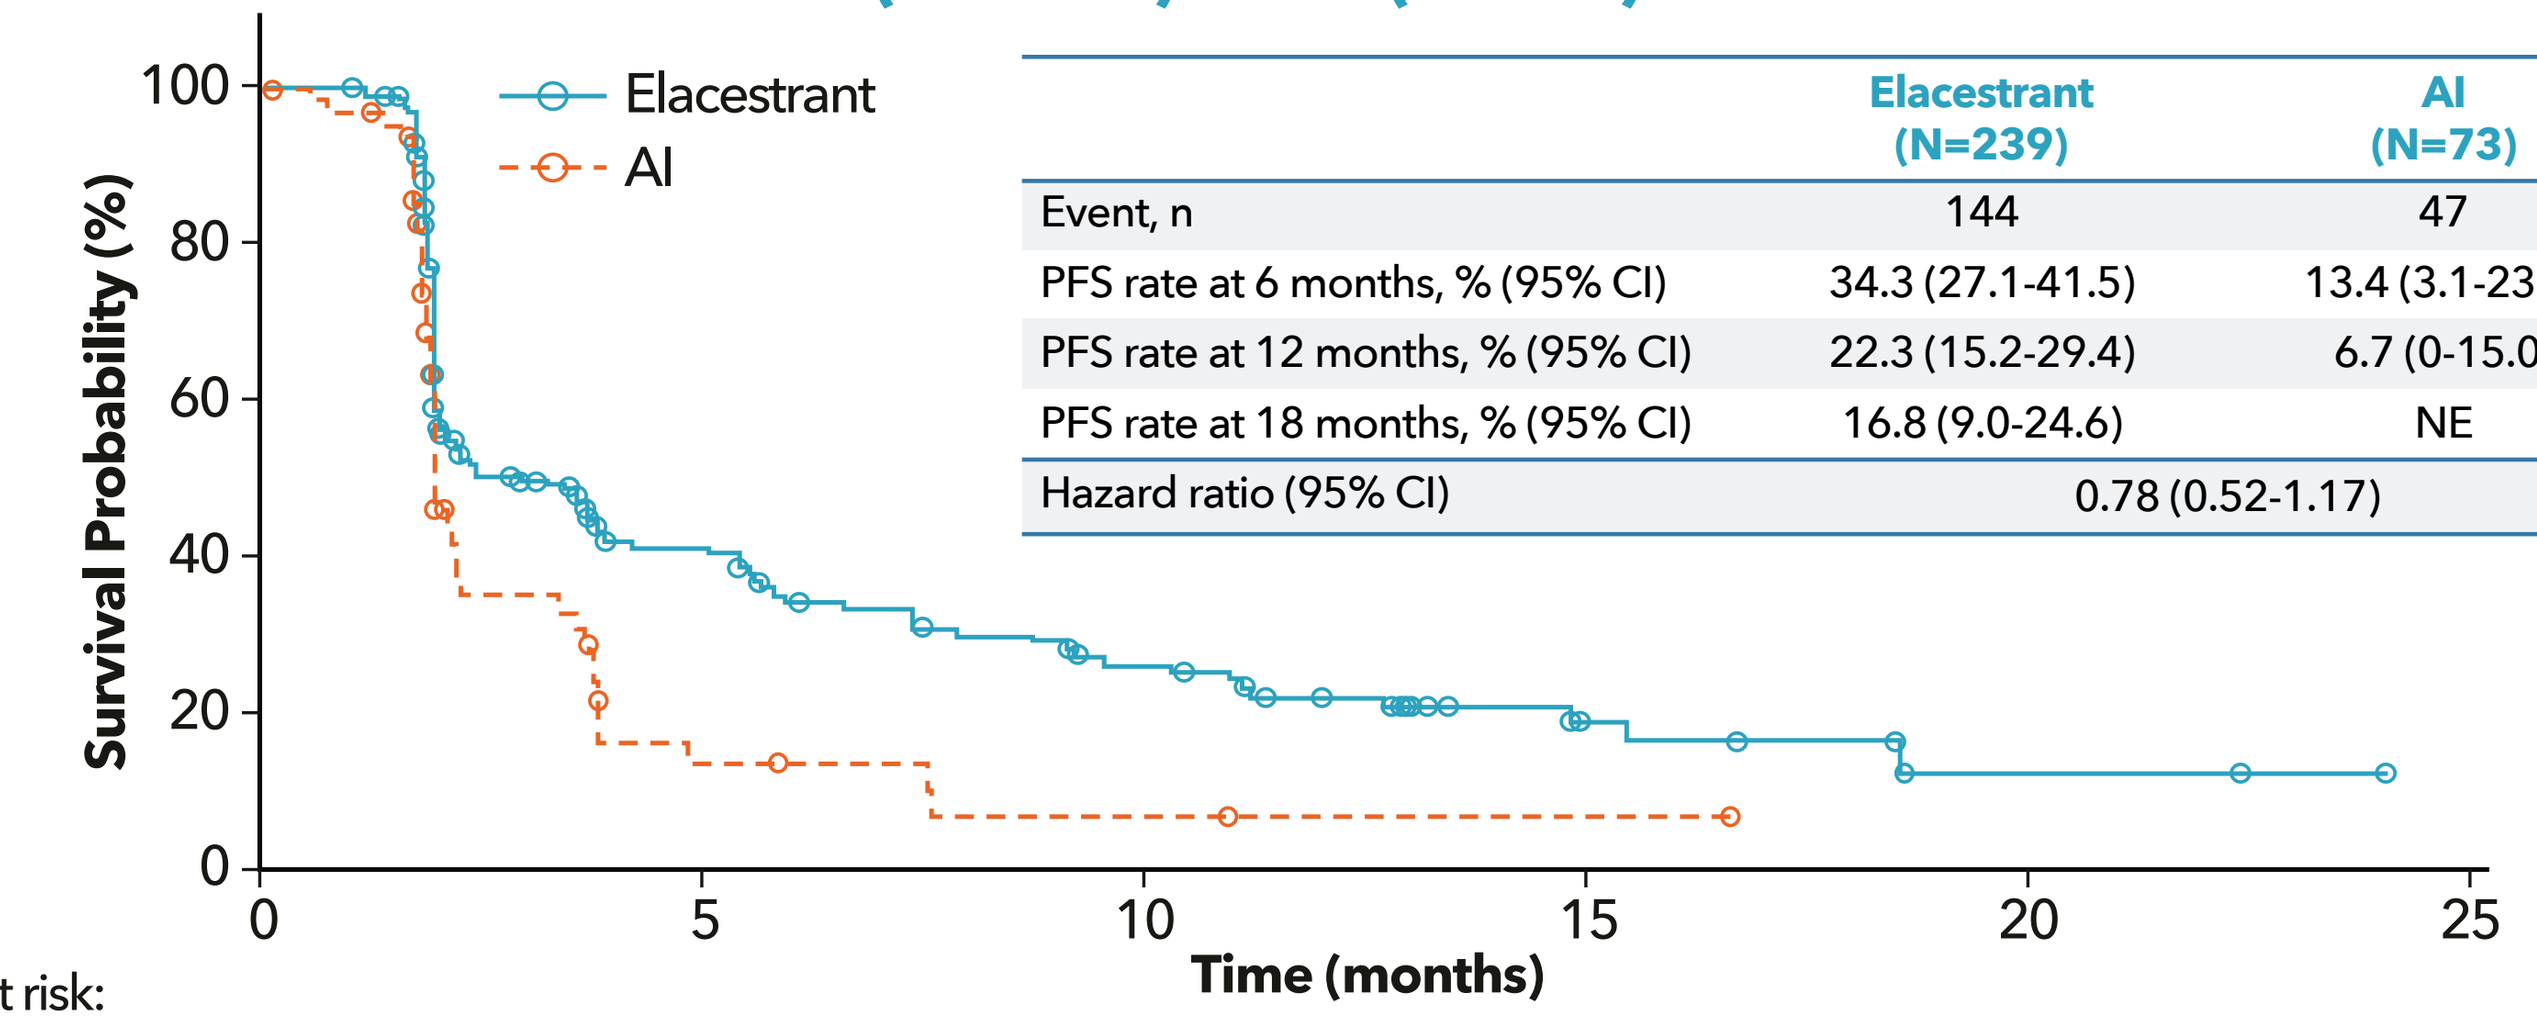
Elacestrant
AI
Survival Probability (%)
100
80
60
40
20
0
0
5
10
15
20
25
Time (months)

	Elacestrant (N=239)	AI (N=73)
Event, n	144	47
PFS rate at 6 months, % (95% CI)	34.3 (27.1-41.5)	13.4 (3.1-23.6)
PFS rate at 12 months, % (95% CI)	22.3 (15.2-29.4)	6.7 (0-15.0)
PFS rate at 18 months, % (95% CI)	16.8 (9.0-24.6)	NE
Hazard ratio (95% CI)	0.78 (0.52-1.17)	

No. at risk:											
Elacestrant	239	92	57	35	27	19	8	6	2	1	0
AIs	73	16	5	2	2	1	1	0			

Elacestrant (N=115) vs Fulvestrant (N=83): Patients With *ESR1*-mut

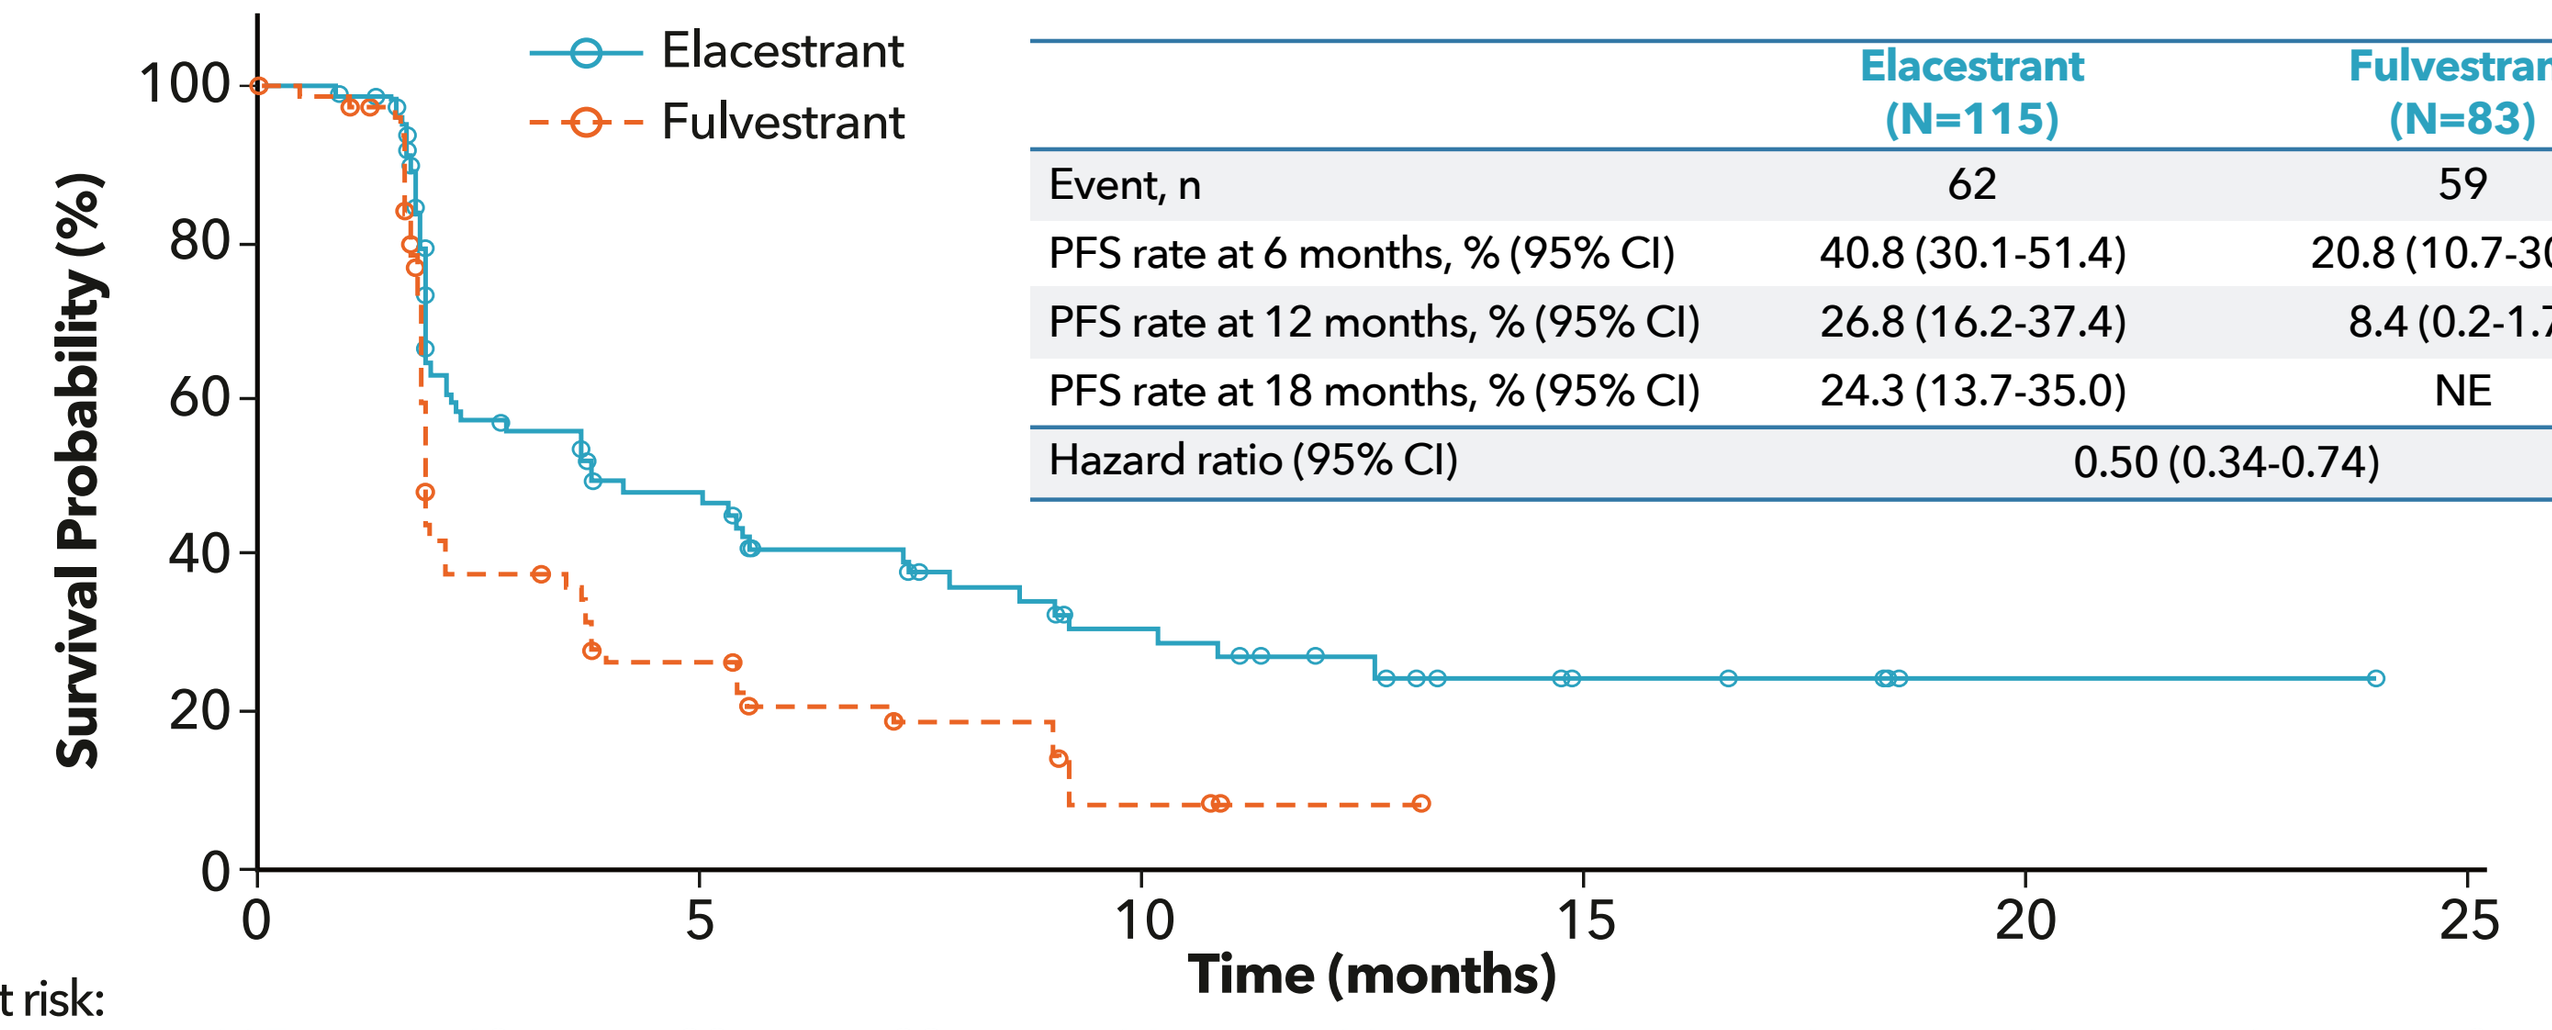
Elacestrant
Fulvestrant
Survival Probability (%)
100
80
60
40
20
0
0
5
10
15
20
25
Time (months)

	Elacestrant (N=115)	Fulvestrant (N=83)
Event, n	62	59
PFS rate at 6 months, % (95% CI)	40.8 (30.1-51.4)	20.8 (10.7-30.8)
PFS rate at 12 months, % (95% CI)	26.8 (16.2-37.4)	8.4 (0.2-1.7)
PFS rate at 18 months, % (95% CI)	24.3 (13.7-35.0)	NE
Hazard ratio (95% CI)	0.50 (0.34-0.74)	

No. at risk:											
Elacestrant	115	48	33	22	16	11	5	4	1	1	0
Fulvestrant	83	25	16	8	3	1	0				

Elacestrant (N=115) vs AI (N=30): Patients With *ESR1*-mut

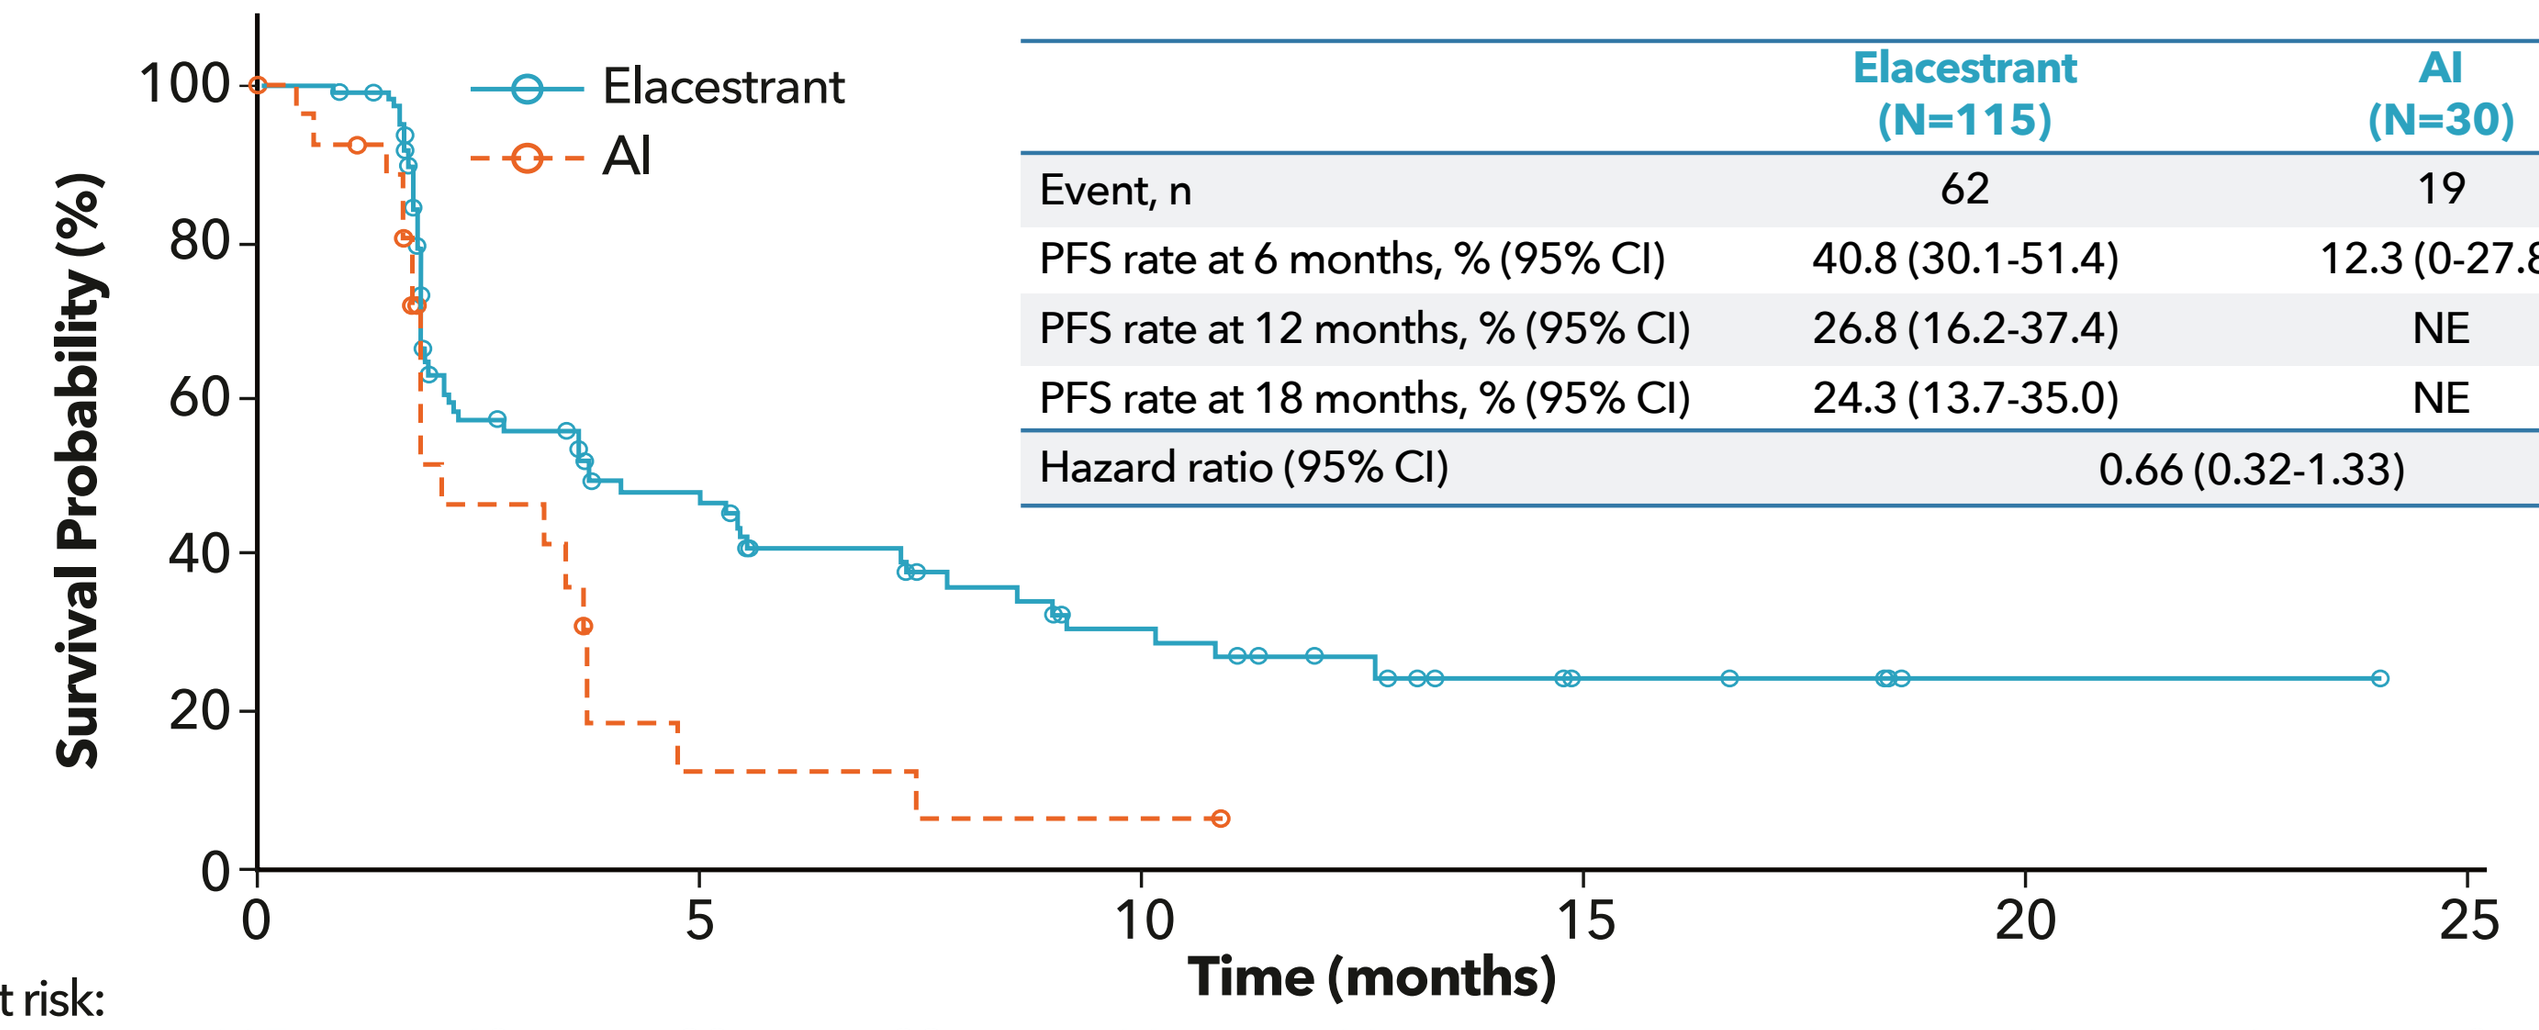
Elacestrant
AI
Survival Probability (%)
100
80
60
40
20
0
0
5
10
15
20
25
Time (months)

	Elacestrant (N=115)	AI (N=30)
Event, n	62	19
PFS rate at 6 months, % (95% CI)	40.8 (30.1-51.4)	12.3 (0-27.8)
PFS rate at 12 months, % (95% CI)	26.8 (16.2-37.4)	NE
PFS rate at 18 months, % (95% CI)	24.3 (13.7-35.0)	NE
Hazard ratio (95% CI)	0.66 (0.32-1.33)	

No. at risk:											
Elacestrant	115	48	33	22	16	11	5	4	1	1	0
AIs	30	9	2	1	1	0					

CI, confidence interval; NE, not evaluable; PFS, progression-free survival.

Safety

- The most common adverse event observed with elacestrant was nausea (**Table 2**).
 - Grade 3 nausea occurred in 2.5%, 0%, and 2.9% of patients receiving elacestrant, fulvestrant, and AI, respectively.
- No Grade 4 events were observed, and there were no treatment-related deaths in either of the groups.
- The incidence of adverse events leading to treatment discontinuation was low in all treatment groups.

Table 2. Adverse Events

Adverse events, n (%)	Elacestrant N=237	SOC		
		All N=229	Fulvestrant N=161	AI N=68
Any treatment-emergent AE	218 (92.0)	197 (86.0)	144 (89.4)	53 (77.9)
Treatment-emergent Grade 3 AE	64 (27.0)	47 (20.5)	33 (20.5)	14 (20.6)
Any treatment-related AE	150 (63.3)	100 (43.7)	72 (44.7)	28 (41.2)
Treatment-related Grade 3 AE	17 (7.2)	7 (3.1)	5 (3.1)	2 (2.9)
Any treatment-related serious AE	3 (1.3)	0	0	0
AE leading to discontinuation of therapy	15 (6.3)	10 (4.4)	6 (3.7)	4 (5.9)
Treatment-emergent AE occurring in >15% of patients[a]				
Nausea	83 (35.0)	43 (18.8)	26 (16.1)	17 (25.0)
Fatigue	45 (19.0)	43 (18.8)	35 (21.7)	8 (11.8)
Vomiting	45 (19.0)	19 (8.3)	12 (7.5)	7 (10.3)
Arthralgia	34 (14.3)	37 (16.2)	28 (17.4)	9 (13.2)

[a]>15% of patients in any treatment group

CONCLUSIONS

- Elacestrant demonstrated statistically significant and clinically meaningful improvement in PFS vs SOC endocrine therapy in a randomized global phase 3 study in patients with ER+/HER2- mBC in the 2nd/3rd-line post-CDK4/6i setting.[1]
- The sequencing guidance provided in the protocol regarding the selection of therapy in the control arm represents real-world therapeutic strategy for patients with ER+/HER2- mBC in the 2nd/3rd-line post-CDK4/6i setting.
- In this post-hoc subgroup analysis, elacestrant improved PFS compared with fulvestrant as well as AI consistently at 6, 12, and 18 months, highlighting superior efficacy of elacestrant regardless of the type of endocrine therapy.
- Elacestrant has a predictable and manageable safety profile consistent with other endocrine therapies.

REFERENCE:
[1]Bidard FC, et al. *J Clin Oncol*. 2022 [published online ahead of print, 2022 May 18]: JCO2200338. doi:10.1200/JCO.22.00338.

ACKNOWLEDGEMENT:
Editorial assistance was provided by Phillips Gilmore Oncology Communications, Inc., and funded by Menarini Group.

Copies of this poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without written permission of the authors. Contact **philippe.aftimos@bordet.be** for permissions and information.